A double-blinded, placebo controlled single ascending dose study to assess safety, tolerability, pharmacokinetics, and pharmacodynamics of a novel long-acting GLP-1/GIP/Glucagon triple agonist (HM15211) in healthy obese subjects.

982-P

Jae Duk Choi[1], Seungjae Baek[1], Youngmin Kim[1], Kahee Eun[1], Se Chang Kwon[1], Linda Morrow[2], Marcus Hompesch[2*]

[1]Hanmi Pharmaceutical Co., Ltd. Seoul, South Korea, [2]ProSciento, Inc. Chula Vista , CA, USA, *Corresponding Author

ABSTRACT

HM15211 is a novel long-acting GLP-1/GIP/Glucagon triple-receptor co-agonist conjugated with a human IgG Fc fragment to extend its half-life. HM15211 has shown therapeutic potential in animal models of NASH and dyslipidemia. This study was a randomized, double-blind, placebo-controlled First-In-Human trial designed to assess safety, pharmacokinetics, and pharmacodynamics of single subcutaneous doses of HM15211 in obese but otherwise healthy adult subjects (41 subjects, mean age 45.7 years, 51.2% males, mean BMI 33.6 kg/m^2). Subjects were randomized to one of 5 sequential ascending doses of either HM15211 or matching placebo. HM15211 was safe and well-tolerated in this population with no significant changes in heart rate, blood pressure, ECG, physical exams, and clinical laboratory tests. There was no subject who discontinued the study due to an adverse event and there were no serious adverse events. Compared to placebo, HM15211 suppressed corresponding endogenous incretins and reduced circulating amino acids levels in a dose dependent manner, which may be related to the mode of action of HM15211. Plasma concentration of HM15211 reached its peak level (T_{max}) in 31.20 to 68.08 hours. Plasma $AUC_{0\text{-}inf}$ and C_{max} increased in a dose-proportional manner and the terminal half-life ($t_{1/2}$) was 72.09 to 142.10 hours. In conclusion, single doses of HM15211 were well tolerated, showed PK profiles suitable for weekly administration, and initial evidence in support of the hypothesized mode of action. Clinical studies in obese NAFLD/NASH patients will follow.

BACKGROUND

HM15211 is a GLP-1/GIP/GCG triple agonist, conjugated with a human IgG Fc fragment *via* a flexible PEG linker

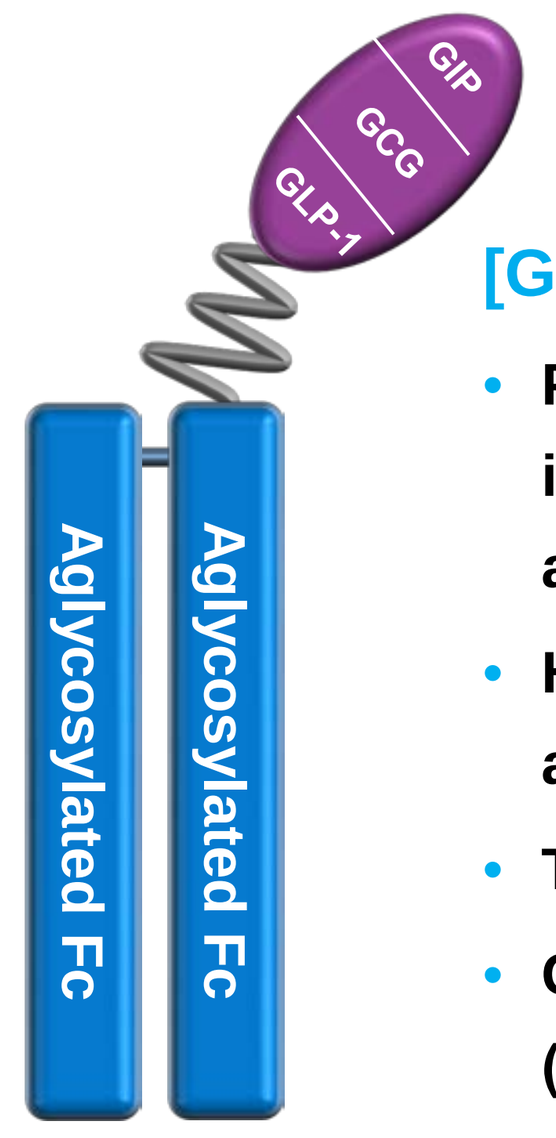

GIP
GCG
GLP-1
Aglycosylated Fc
Aglycosylated Fc

[General Profile of HM15211]

- **Potential to improve lipid profiles, and anti-inflammatory effects through the addition of GIP activity**
- **High glucagon activity favors liver preferential activity and maybe advantageous for NASH treatment**
- **The extended half life is sufficient for weekly dosing**
- **Good Solubility (≥ 150 mg/mL) & bioavailability (≥ 95 %)**

STUDY DESIGN

Figure 1. Study Design

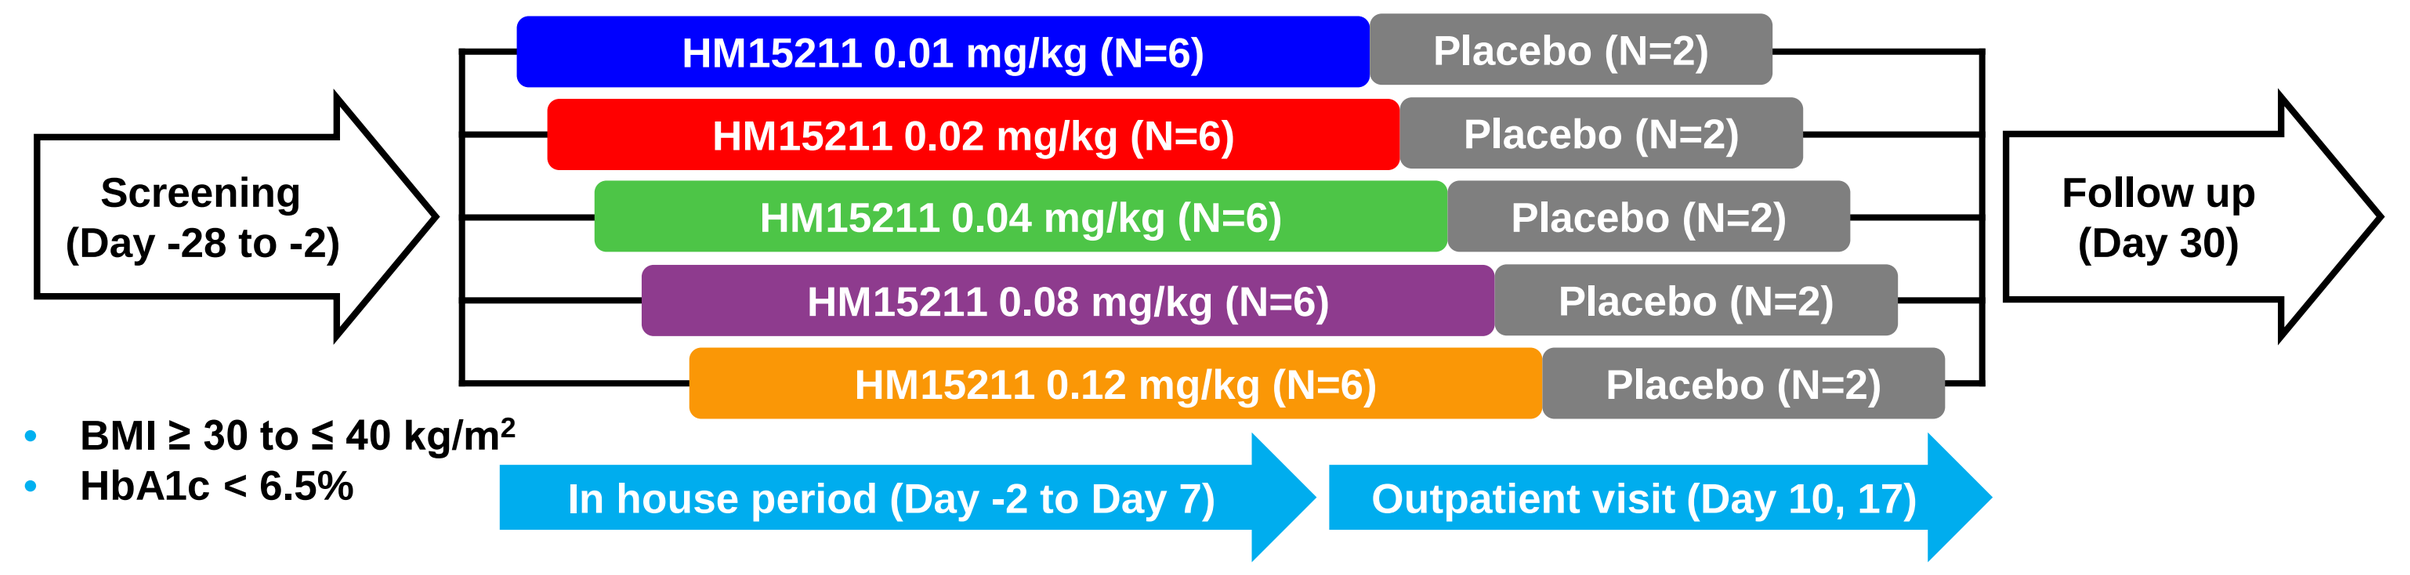

Screening (Day -28 to -2)
HM15211 0.01 mg/kg (N=6)
HM15211 0.02 mg/kg (N=6)
HM15211 0.04 mg/kg (N=6)
HM15211 0.08 mg/kg (N=6)
HM15211 0.12 mg/kg (N=6)
Placebo (N=2)
Follow up (Day 30)
BMI ≥ 30 to ≤ 40 kg/m2
HbA1c < 6.5%
In house period (Day -2 to Day 7)
Outpatient visit (Day 10, 17)

Study Objectives

- **Primary objectives**
 - To assess safety and tolerability of HM15211 after single subcutaneous (SC) doses
 - To assess phamacokinetic (PK) profiles of HM15211
- **Secondary objectives**
 - To assess pharmacodynamic (PD) properties of HM15211

RESULTS

Table 1. Baseline Characteristics

	HM15211					Placebo
	0.01 mg/kg (N=6)	0.02 mg/kg (N=6)	0.04 mg/kg (N=6)	0.08 mg/kg (N=7)	0.12 mg/kg (N=6)	(N=10)
Ages: years (SD)	41.0 (14.3)	36.3 (11.8)	54.0 (6.1)	49.7 (11.1)	43.8 (13.5)	47.4 (6.6)
Sex: M/F (%)	50/50	33/67	67/33	43/57	67/33	50/50
Race (%)						
White	4 (67)	5 (83)	2 (33)	5 (71)	6 (100)	5 (50)
Black or African American	2 (33)	1 (17)	4 (67)	1 (14)	0 (0)	4 (40)
Asian	0 (0)	0 (0)	0 (0)	0 (0)	0 (0)	1 (10)
Native Hawaiian or Other Pacific Islander	0 (0)	0 (0)	0 (0)	1 (14)	0 (0)	0 (0)
Weight: kg (SD)	95.7 (8.5)	97.1 (16.8)	96.9 (10.3)	92.6 (11.0)	87.3 (14.5)	96.0 (10.7)
BMI: kg/m^2 (SD)	33.2 (1.7)	34.9 (3.4)	33.1 (2.4)	32.7 (1.8)	32.7 (2.6)	34.5 (3.6)

- One withdrawn subject in cohort 4 was replaced

Figure 2. Serum PK Exposure of HM15211 (semi-log)

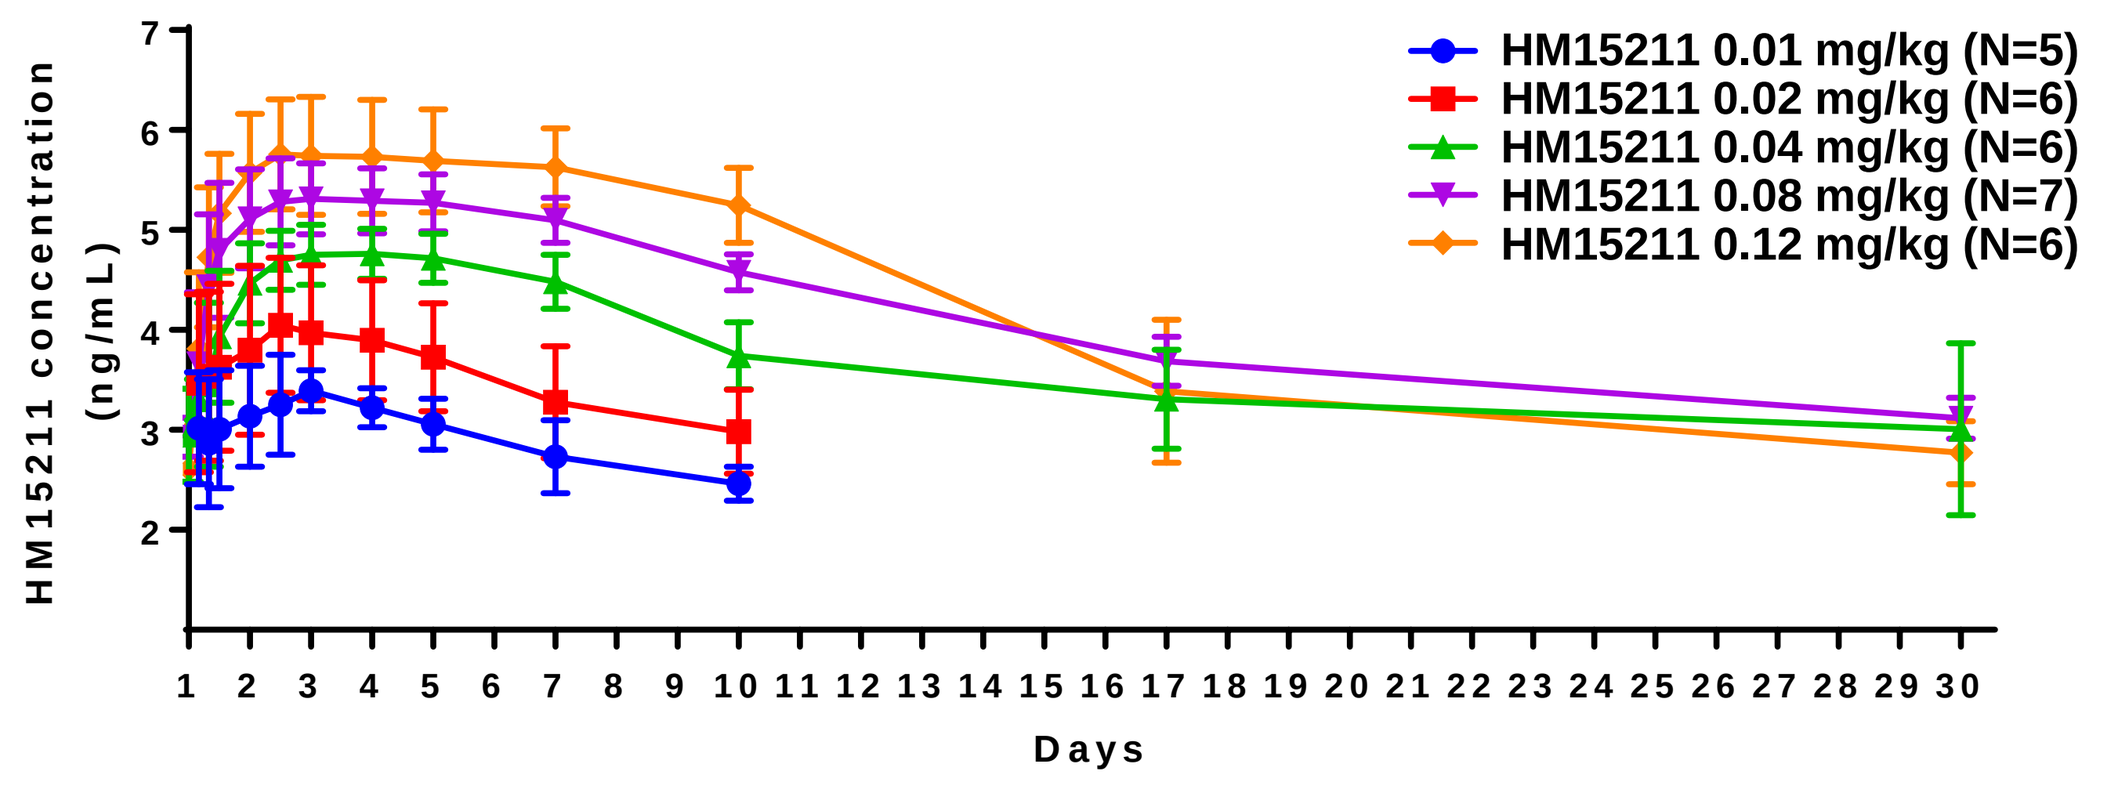

HM15211 concentration (ng/mL)
Days
HM15211 0.01 mg/kg (N=5)
HM15211 0.02 mg/kg (N=6)
HM15211 0.04 mg/kg (N=6)
HM15211 0.08 mg/kg (N=7)
HM15211 0.12 mg/kg (N=6)

- Presented data are mean with standard deviation (SD)
- A subject in cohort 1, whose concentration is BLQ at all time-points, are excluded in the analysis

Table 2. Pharmacokinetic parameters

	HM15211 0.01 mg/kg (N=5)	HM15211 0.02 mg/kg (N=6)	HM15211 0.04 mg/kg (N=6)	HM15211 0.08 mg/kg (N=7)	HM15211 0.12 mg/kg (N=6)
C_{max} (ng/mL)	29.76 (14.707)	70.52 (39.699)	127.52 (34.227)	232.14 (82.495)	383.67 (185.738)
T_{max} (hr)	31.20 (10.733)	42.03 (16.527)	66.01 (25.996)	61.71 (40.652)	68.08 (43.316)
$T_{1/2}$ (hr)	72.09 (44.939)	77.24 (26.196)	89.26 (20.012)	142.10 (54.641)	79.18 (19.989)
$AUC_{0\text{-}inf}$ (ng/mL·h)	5034.63 (1322.73)	10189.05 (4584.02)	24603.83 (8530.78)	52575.52 (5808.64)	90903.07 (34897.44)
Dose-normalized C_{max} (ng/mL/mg)	30.73 (13.34)	35.99 (21.21)	33.13 (9.70)	31.46 (11.00)	38.39 (23.25)
Dose-normalized AUC_{inf} (ng/mL·h/mg)	5899.11 (1373.98)	5099.07 (1912.91)	8021.05 (3986.35)	8887.11 (4052.88)	8718.46 (3084.28)

- Parameters are mean with standard deviation (SD)

Table 3. Summary of Adverse Events

TEAE Category / No. of Subjects (%)	HM15211 0.01 mg/kg (N=6)	HM15211 0.02 mg/kg (N=6)	HM15211 0.04 mg/kg (N=6)	HM15211 0.08 mg/kg (N=7)	HM15211 0.12 mg/kg (N=6)	Placebo (N=10)
Any TEAE	3 (50.0)	2 (33.3)	3 (50.0)	7 (100.0)	4 (66.7)	7 (70.0)
Maximum Severity of TEAE						
Mild	3 (50.0)	2 (33.3)	3 (50.0)	6 (85.7)	3 (50.0)	7 (70.0)
Moderate	0 (0.0)	0 (0.0)	0 (0.0)	1[a] (14.3)	1[b] (16.7)	0 (0.0)
Severe	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)
Any Serious TEAE	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)
Any TEAE Leading to Study Discontinuation	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)
Any TEAE Related to Study Medication	0 (0.0)	0 (0.0)	0 (0.0)	5 (71.4)	4 (66.7)	1 (10.0)
Gastrointestinal disorders	0 (0.0)	0 (0.0)	1 (16.7)	4 (57.1)	4 (66.7)	1 (10.0)
Abdominal distension	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	1 (16.7)	0 (0.0)
Abdominal pain	0 (0.0)	0 (0.0)	0 (0.0)	1 (14.3)	1 (16.7)	0 (0.0)
Abdominal pain upper	0 (0.0)	0 (0.0)	0 (0.0)	1 (14.3)	0 (0.0)	0 (0.0)
Abdominal tenderness	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	1 (10.0)
Diarrhea	0 (0.0)	0 (0.0)	0 (0.0)	1 (14.3)	0 (0.0)	0 (0.0)
Dyspepsia	0 (0.0)	0 (0.0)	1 (16.7)	0 (0.0)	0 (0.0)	0 (0.0)
Nausea	0 (0.0)	0 (0.0)	0 (0.0)	4 (57.1)	4 (66.7)	0 (0.0)
Vomiting	0 (0.0)	0 (0.0)	0 (0.0)	2 (28.6)	2 (33.3)	0 (0.0)

- Most of AEs were mild, and all of GI events were observed at the highest two doses (HM15211 0.08 and 0.12 mg/kg) including each one of moderate case of nausea (a) and vomiting (b), respectively

Figure 3. Gastrointestinal Events over Time (Day)

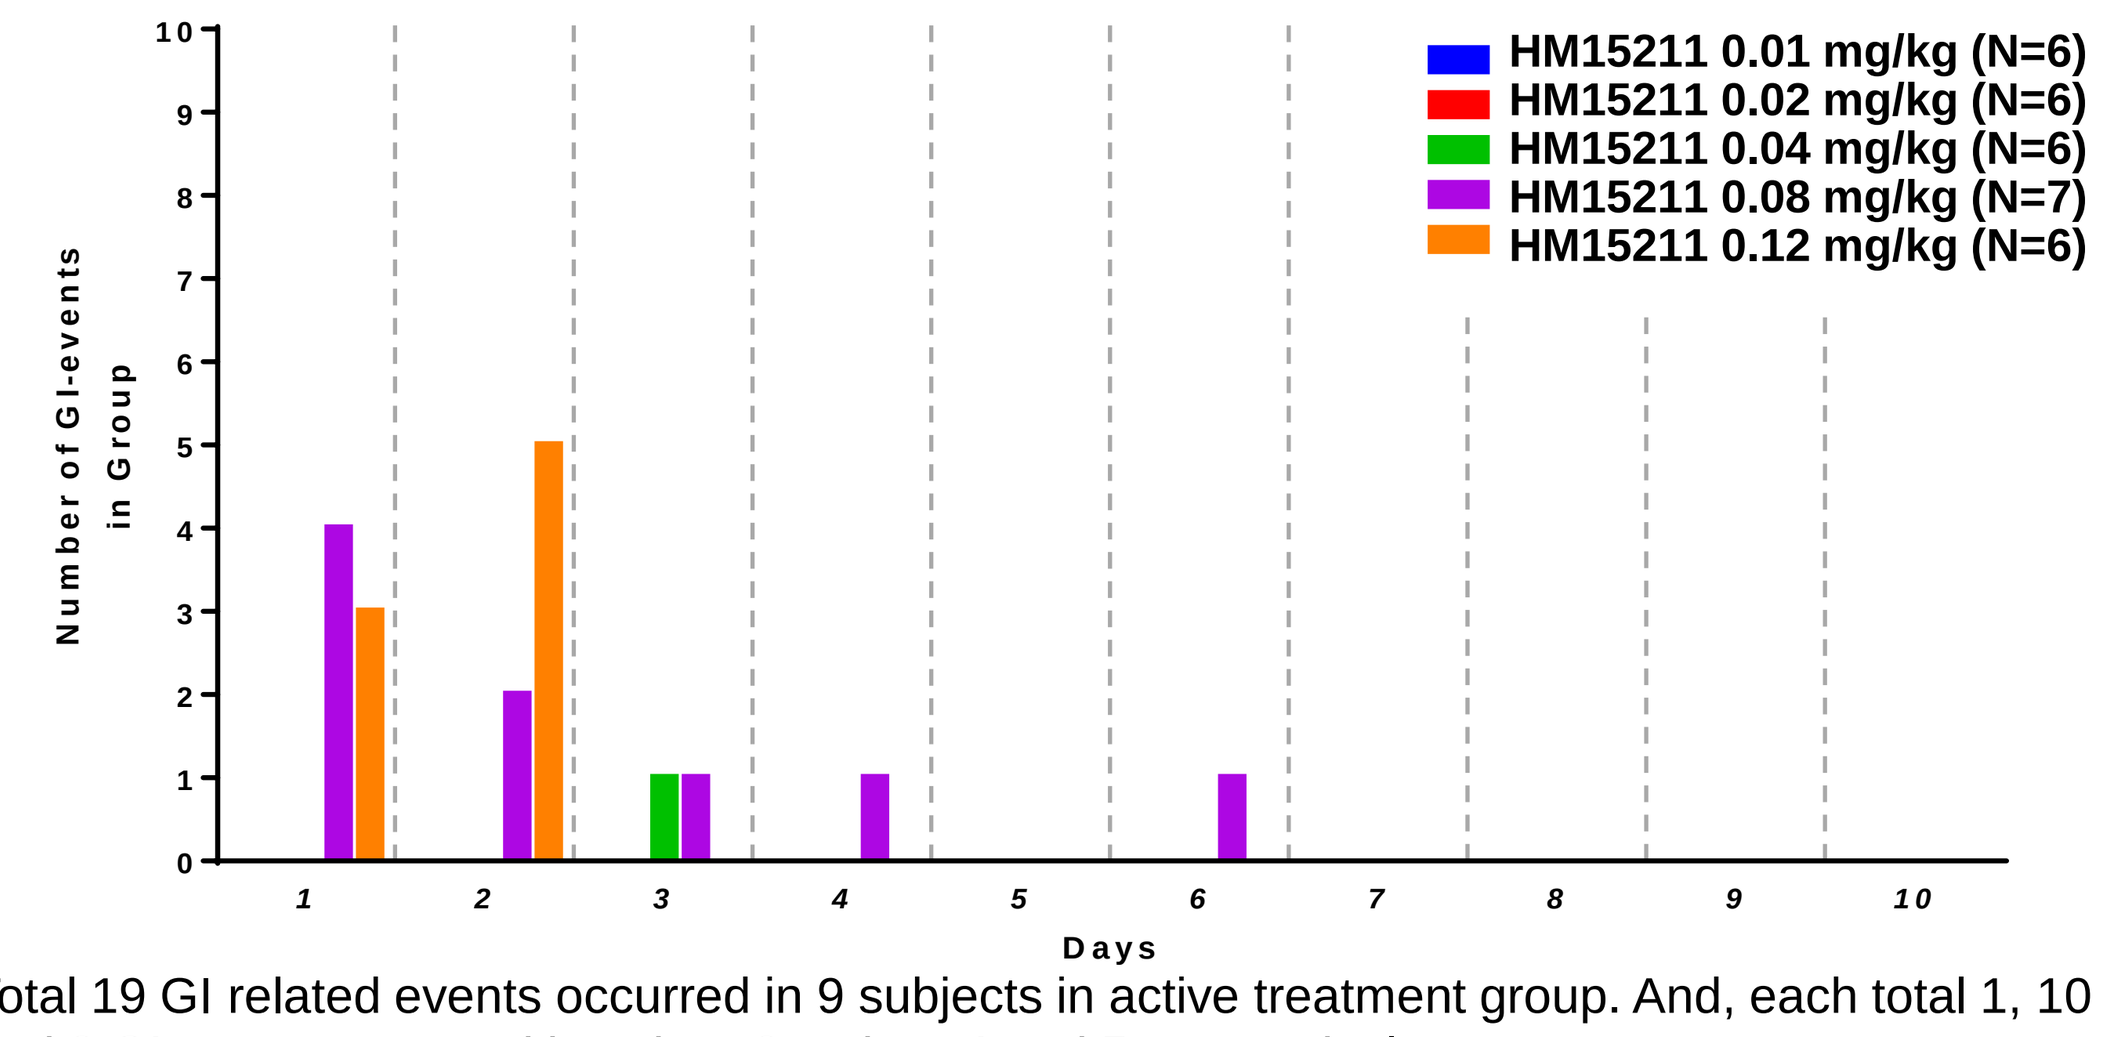

Number of GI-events in Group
Days
HM15211 0.01 mg/kg (N=6)
HM15211 0.02 mg/kg (N=6)
HM15211 0.04 mg/kg (N=6)
HM15211 0.08 mg/kg (N=7)
HM15211 0.12 mg/kg (N=6)

- Total 19 GI related events occurred in 9 subjects in active treatment group. And, each total 1, 10 and 8 GI events occurred in cohort 3, cohort 4 and 5, respectively
- The most frequent gastrointestinal events were nausea and vomiting. Each cases occurred were indicated as dyspepsia and abdominal pain upper on Day 3 and diarrhea and abdominal pain on Day 4 and 6

Figure 4. 24 hr Ambulatory Monitoring: Heart Rate and Systolic/Diastolic Blood Pressure at peak PK Concentration

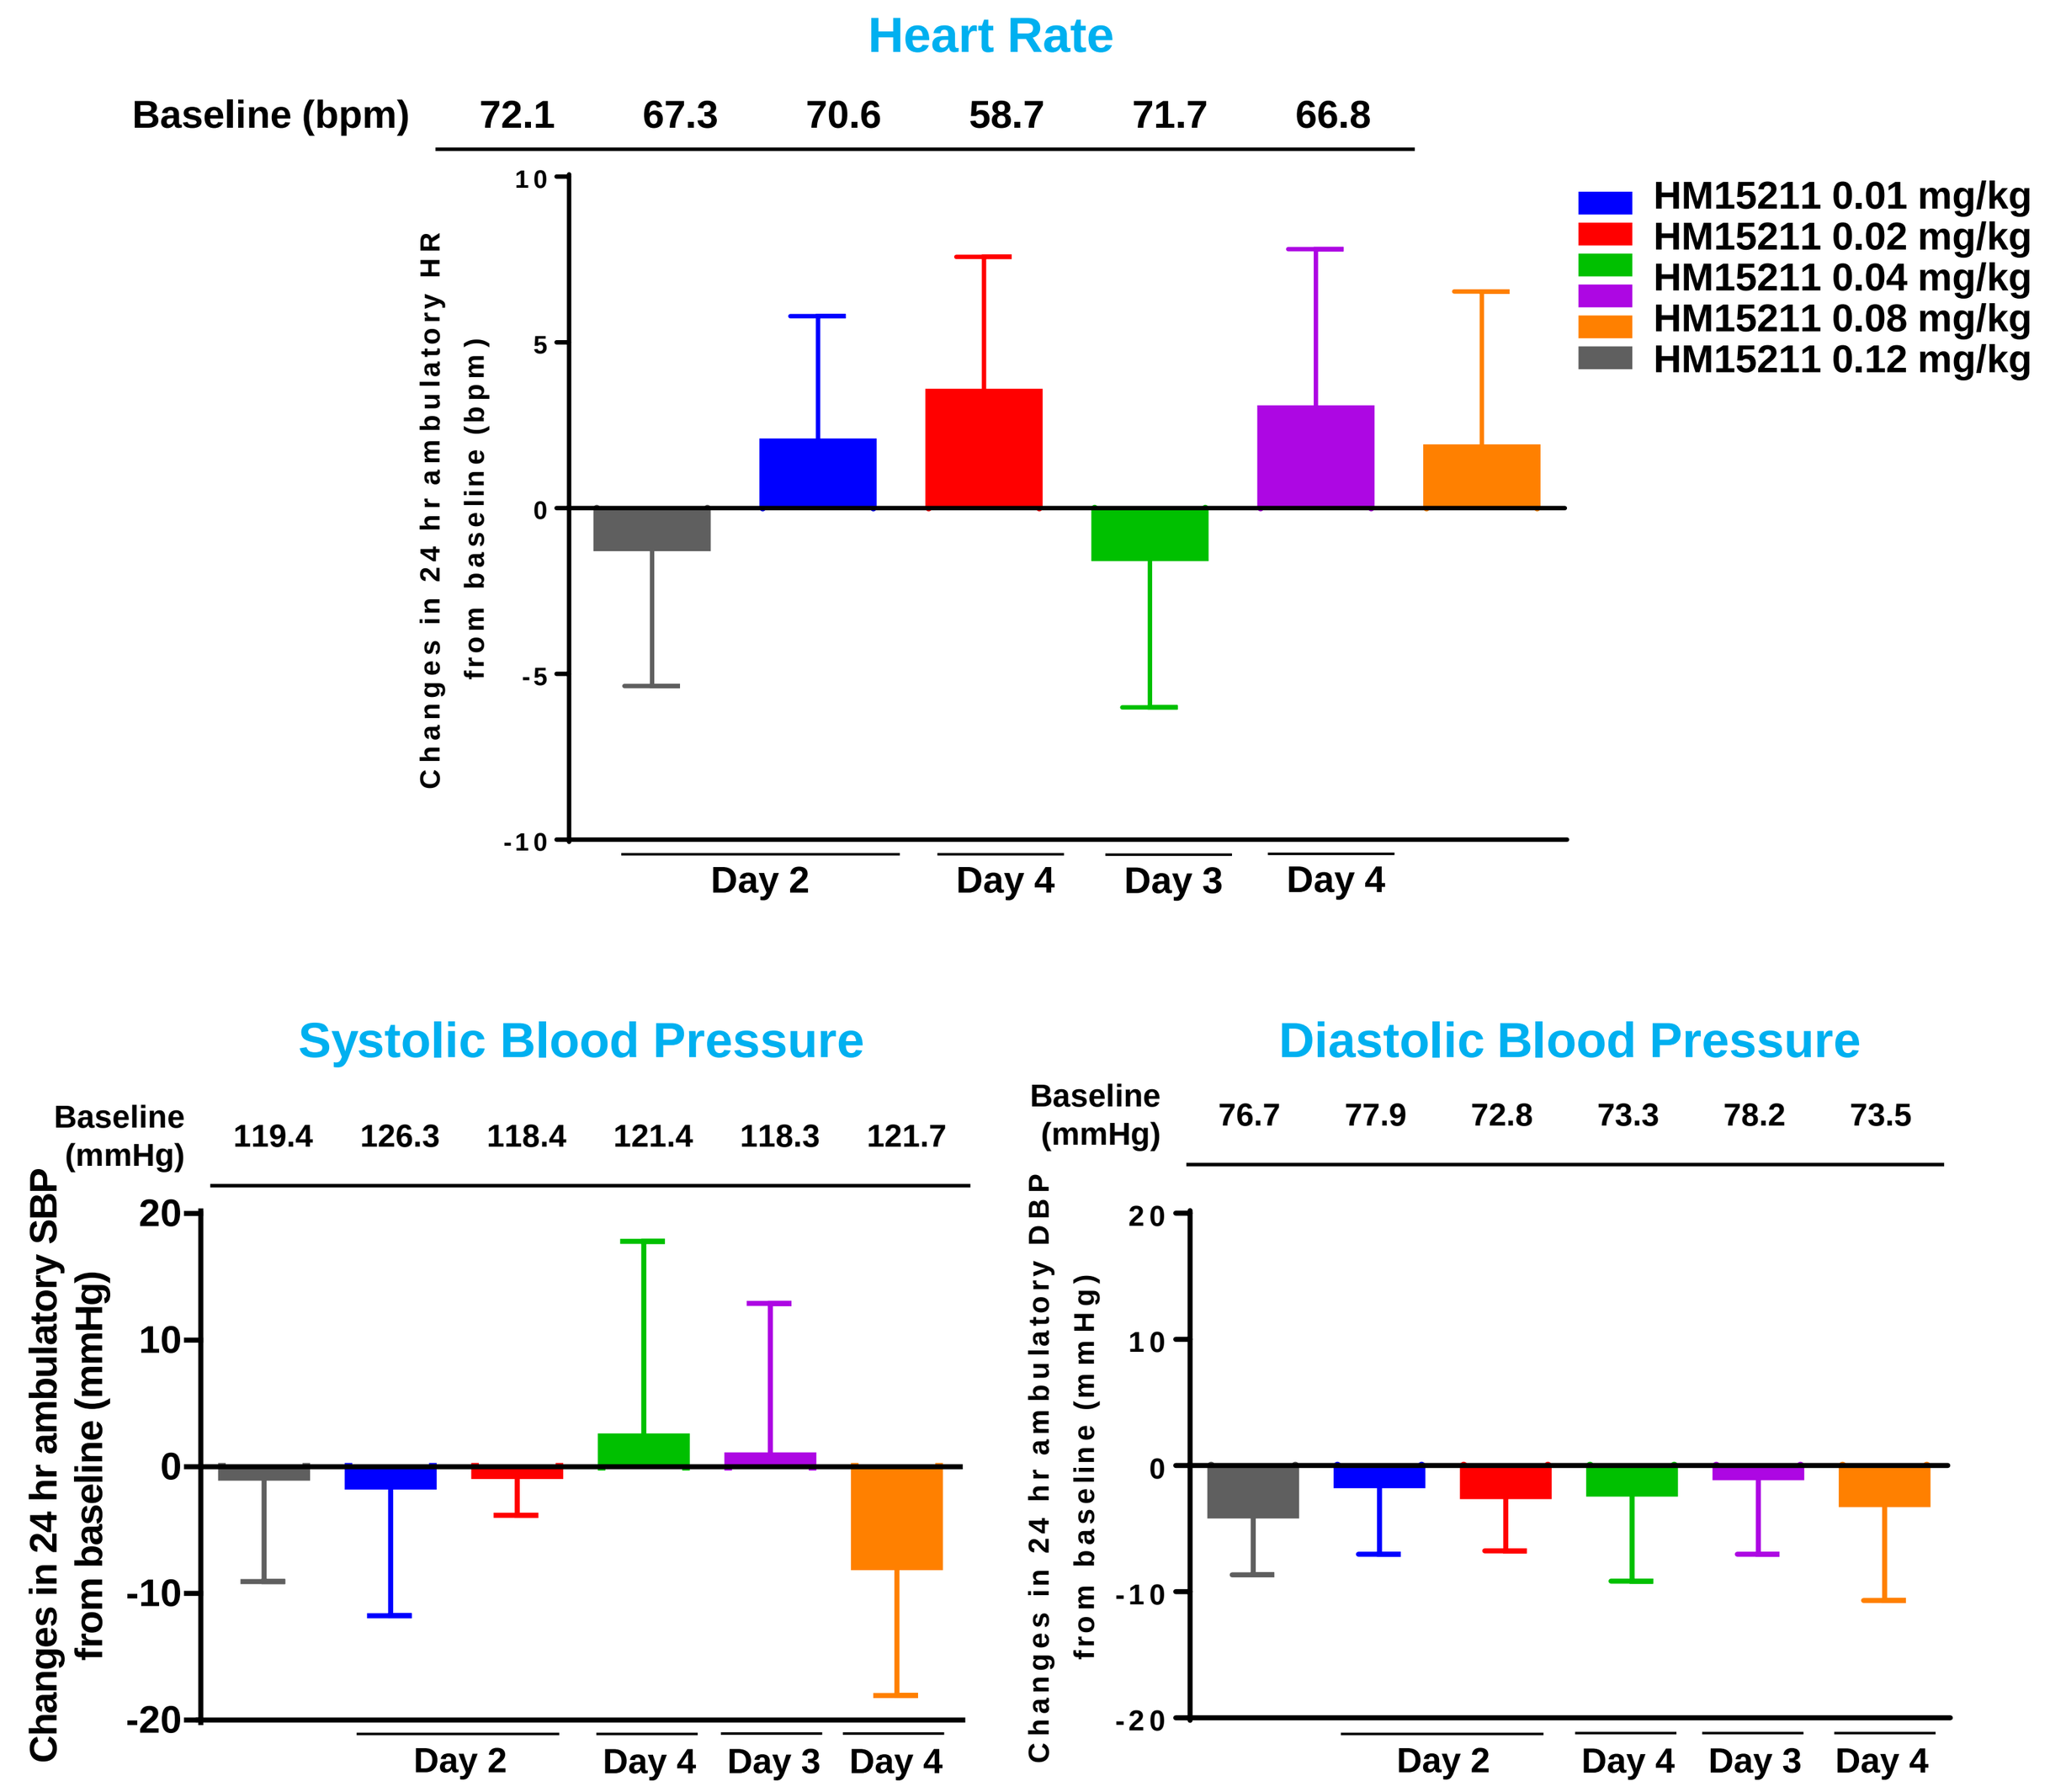

Heart Rate
Baseline (bpm) 72.1 67.3 70.6 58.7 71.7 66.8
Changes in 24 hr ambulatory HR from baseline (bpm)
Day 2 Day 4 Day 3 Day 4
HM15211 0.01 mg/kg
HM15211 0.02 mg/kg
HM15211 0.04 mg/kg
HM15211 0.08 mg/kg
HM15211 0.12 mg/kg
Systolic Blood Pressure
Baseline (mmHg) 119.4 126.3 118.4 121.4 118.3 121.7
Changes in 24 hr ambulatory SBP from baseline (mmHg)
Diastolic Blood Pressure
Baseline (mmHg) 76.7 77.9 72.8 73.3 78.2 73.5
Changes in 24 hr ambulatory DBP from baseline (mmHg)
Day 2 Day 4 Day 3 Day 4

- Presented data are mean with standard deviation (SD)
- Heart rate and Blood Pressure at peak drug concentration were assessed on Day 2 for cohort 1 and 2, and on Day 4 for cohort 3 and 5, and on Day 3 for cohort 4

Table 4. Change from Baseline (24 hr Ambulatory Monitoring)

	HM15211 0.01 mg/kg	HM15211 0.02 mg/kg	HM15211 0.04 mg/kg	HM15211 0.08 mg/kg	HM15211 0.12 mg/kg	Placebo
HR (bpm)	2.0 (3.5)	3.3 (4.1)	-1.5 (4.4)	3.3 (4.8)	1.8 (4.7)	0.4 (4.2)
SPB (mmHg)	-1.5 (10.4)	-0.7 (3.1)	2.3 (15.4)	0.7 (12.3)	-7.9 (10.3)	0.5 (8.2)
DBP (mmHg)	-1.5 (5.6)	-2.3 (4.3)	-2.2 (6.9)	-0.6 (6.3)	-2.9 (7.9)	-3.3 (4.7)
RPP (bpm x mmHg)	146.5 (802.8)	336.5 (661.1)	-123.5 (567.1)	478.0 (936.3)	-299.6 (1090.6)	185.3 (987.1)

- RPP: Rate pressure product
- Presented data are mean with standard deviation (SD)
- Heart rate and Blood Pressure at peak drug concentration were assessed on Day 2 for cohort 1 and 2, and on Day 4 for cohort 3 and 5, and on Day 3 for cohort 4

Figure 5. Incretins and Glucagon at peak PK Concentration

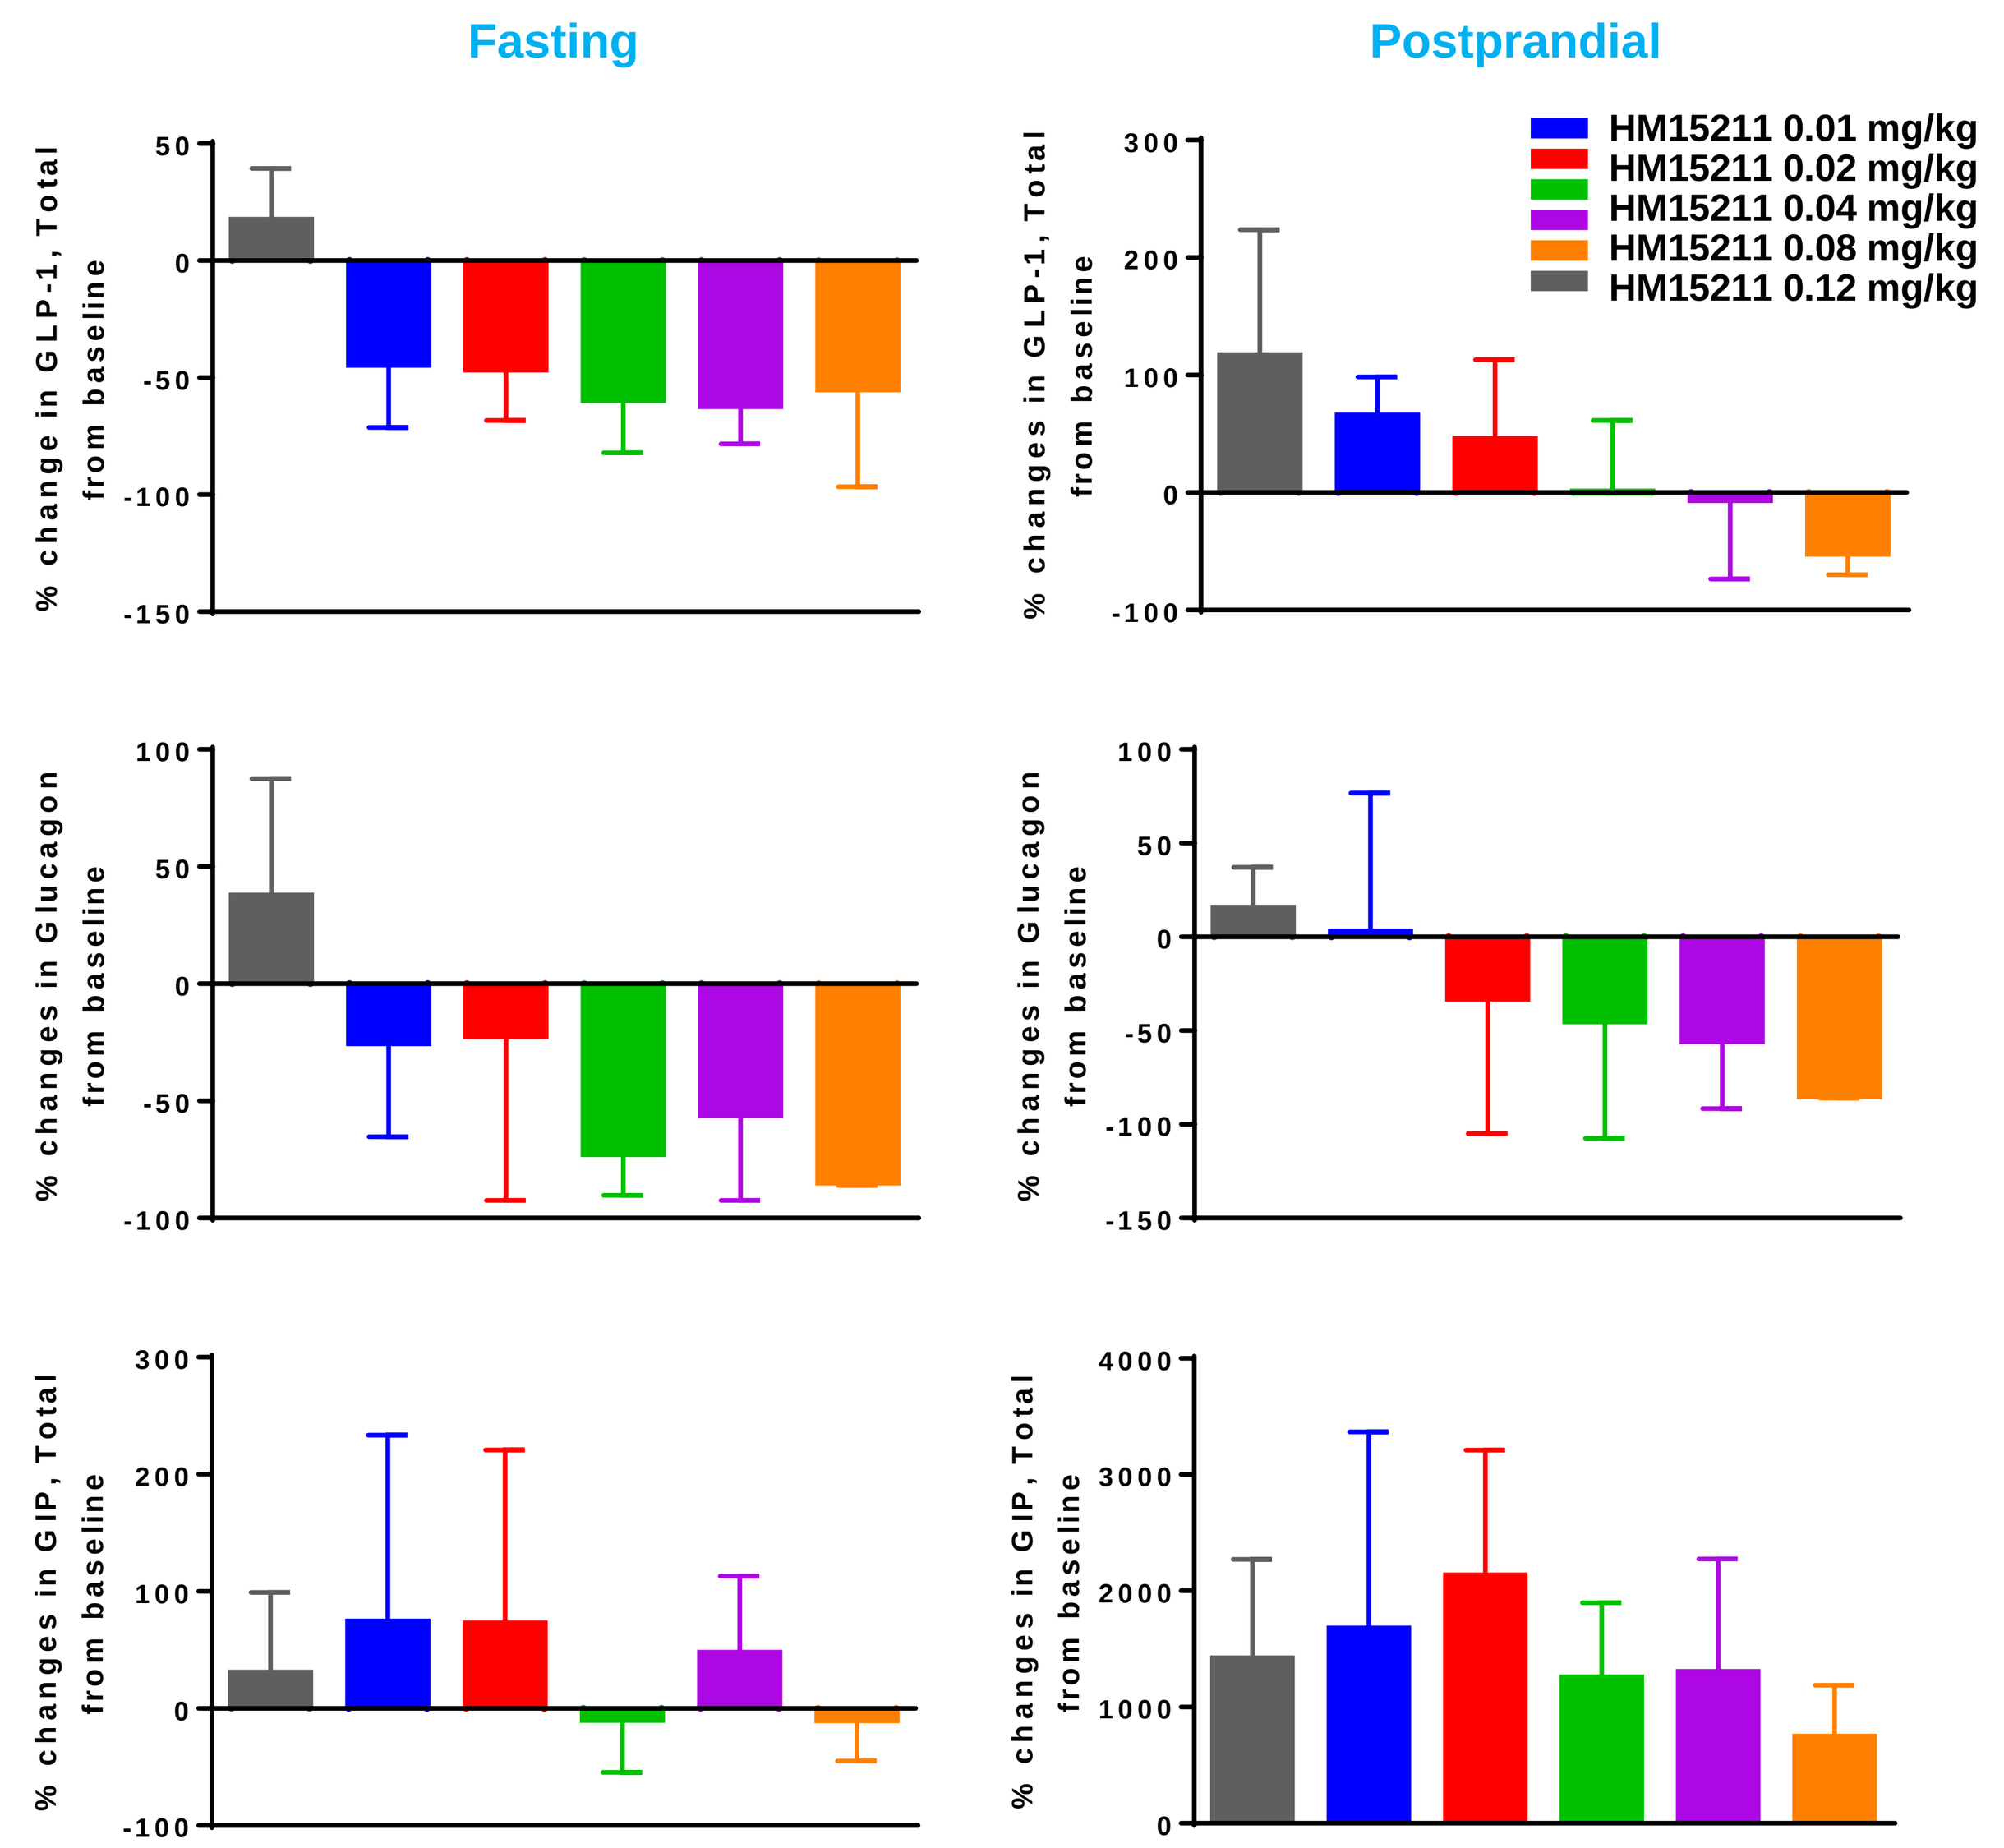

Fasting
Postprandial
% change in GLP-1, Total from baseline
% changes in GLP-1, Total from baseline
% changes in Glucagon from baseline
% changes in GIP, Total from baseline
HM15211 0.01 mg/kg
HM15211 0.02 mg/kg
HM15211 0.04 mg/kg
HM15211 0.08 mg/kg
HM15211 0.12 mg/kg

- Presented data are mean with standard deviation (SD)
- Incretin levels were assessed on 48 hours after dosing for fasting and 36 hours after for postprandial

CONCLUSIONS

- **HM15211 was safe and well tolerated after a single SC injection in healthy subjects**
- **The PK profiles of up to 0.12 mg/kg subcutaneous injection of HM15211 showed a dose dependent increase in circulating HM15211 and the potential for weekly dosing**
- **The reduction in incretins and glucagon confirmed the hypothesized effects of HM15211 on those biomarkers**
- **A Phase 1, 12-week multiple ascending dose study to establish safety, MTD and initial PD is currently ongoing in obese NAFLD subjects**

American Diabetes Association's (ADA) 79th Scientific Sessions, San Francisco, CA, USA; June 07-11, 2019

Hanmi Pharm. Co., Ltd.