Therapeutic effect of a long-acting GLP-1/GIP/Glucagon triple agonist (HM15211) in a dyslipidemia animal model

996-P

Jae Hyuk Choi[1], Hyo Sang Jo[1], Jung Kuk Kim[1], Sang Don Lee[1], Jong Soo Lee[1], Sang-Hyun Lee[1], In Young Choi[1]

[1]Hanmi Pharm. Co., Ltd, Seoul, South Korea

ABSTRACT

Dyslipidemia is a well-established risk factor for cardiovascular disease. Although an efficacy has been demonstrated for several drugs including HMGCR inhibitor and PCSK9 inhibitor, overall outcome was still in doubt due to heterogeneity of this disease. Thus, targeting multiple pathways may provide promising efficacy in broad patient population. Previously, we showed that a long-acting GLP-1/GIP/Glucagon triple agonist, HM15211, not only provided efficient weight loss, but also improved lipid profiles in DIO mice. Of note, unlike current drugs possessing a single target, HM15211 could affect multiple pathways in lipid metabolism, suggesting HM15211 as a potential therapeutic option for dyslipidemia. Here, we investigated the effect of HM15211 on dyslipidemia and its modes of action (MoAs)

In high fat- and high fructose-fed hamsters, HM15211 showed greater LDL lowering than commercial drugs (-85.3% for HM15211, -18.1% and -32.8% for evolocumab and rosuvastatin, respectively *vs.* Veh.). To elucidate the responsible MoAs, potential effects of HM15211 on lipid absorption was evaluated in normal mice, and HM15211 showed significantly decreased plasma TG level after oral lipid challenge, compared to commercial drugs. Next, HepG2 cells were treated with HM15211, and substantially increased LDLR expression and more LDL uptake than evolocumab (3.6-fold for HM15211, 2.8-fold for evolocumab *vs.* CTL) were observed. To explore the additional HM15211 targets, the activity status of HMGCR, the key enzyme for cholesterol synthesis, was evaluated. Surprisingly, HM15211 induced progressive degradation of HMGCR both in HepG2 cells and hamster liver tissue, thereby substantially inhibiting its enzymatic activity. Furthermore, HM15211 led to a favorable gene expression signature associated with free-fatty acid (FFA) metabolism. Increased ApoA1 and ABCA1 expression may explain, at least in part, the increase trend of HDL by HM15211

In conclusion, we propose that by targeting multiple steps essential for LDL and FFA metabolism, HM15211 could provide therapeutic benefits for dyslipidemia patients

BACKGROUND

Known targets of current dyslipidemia drugs, and suggested effects of HM15211 on lipid metabolism

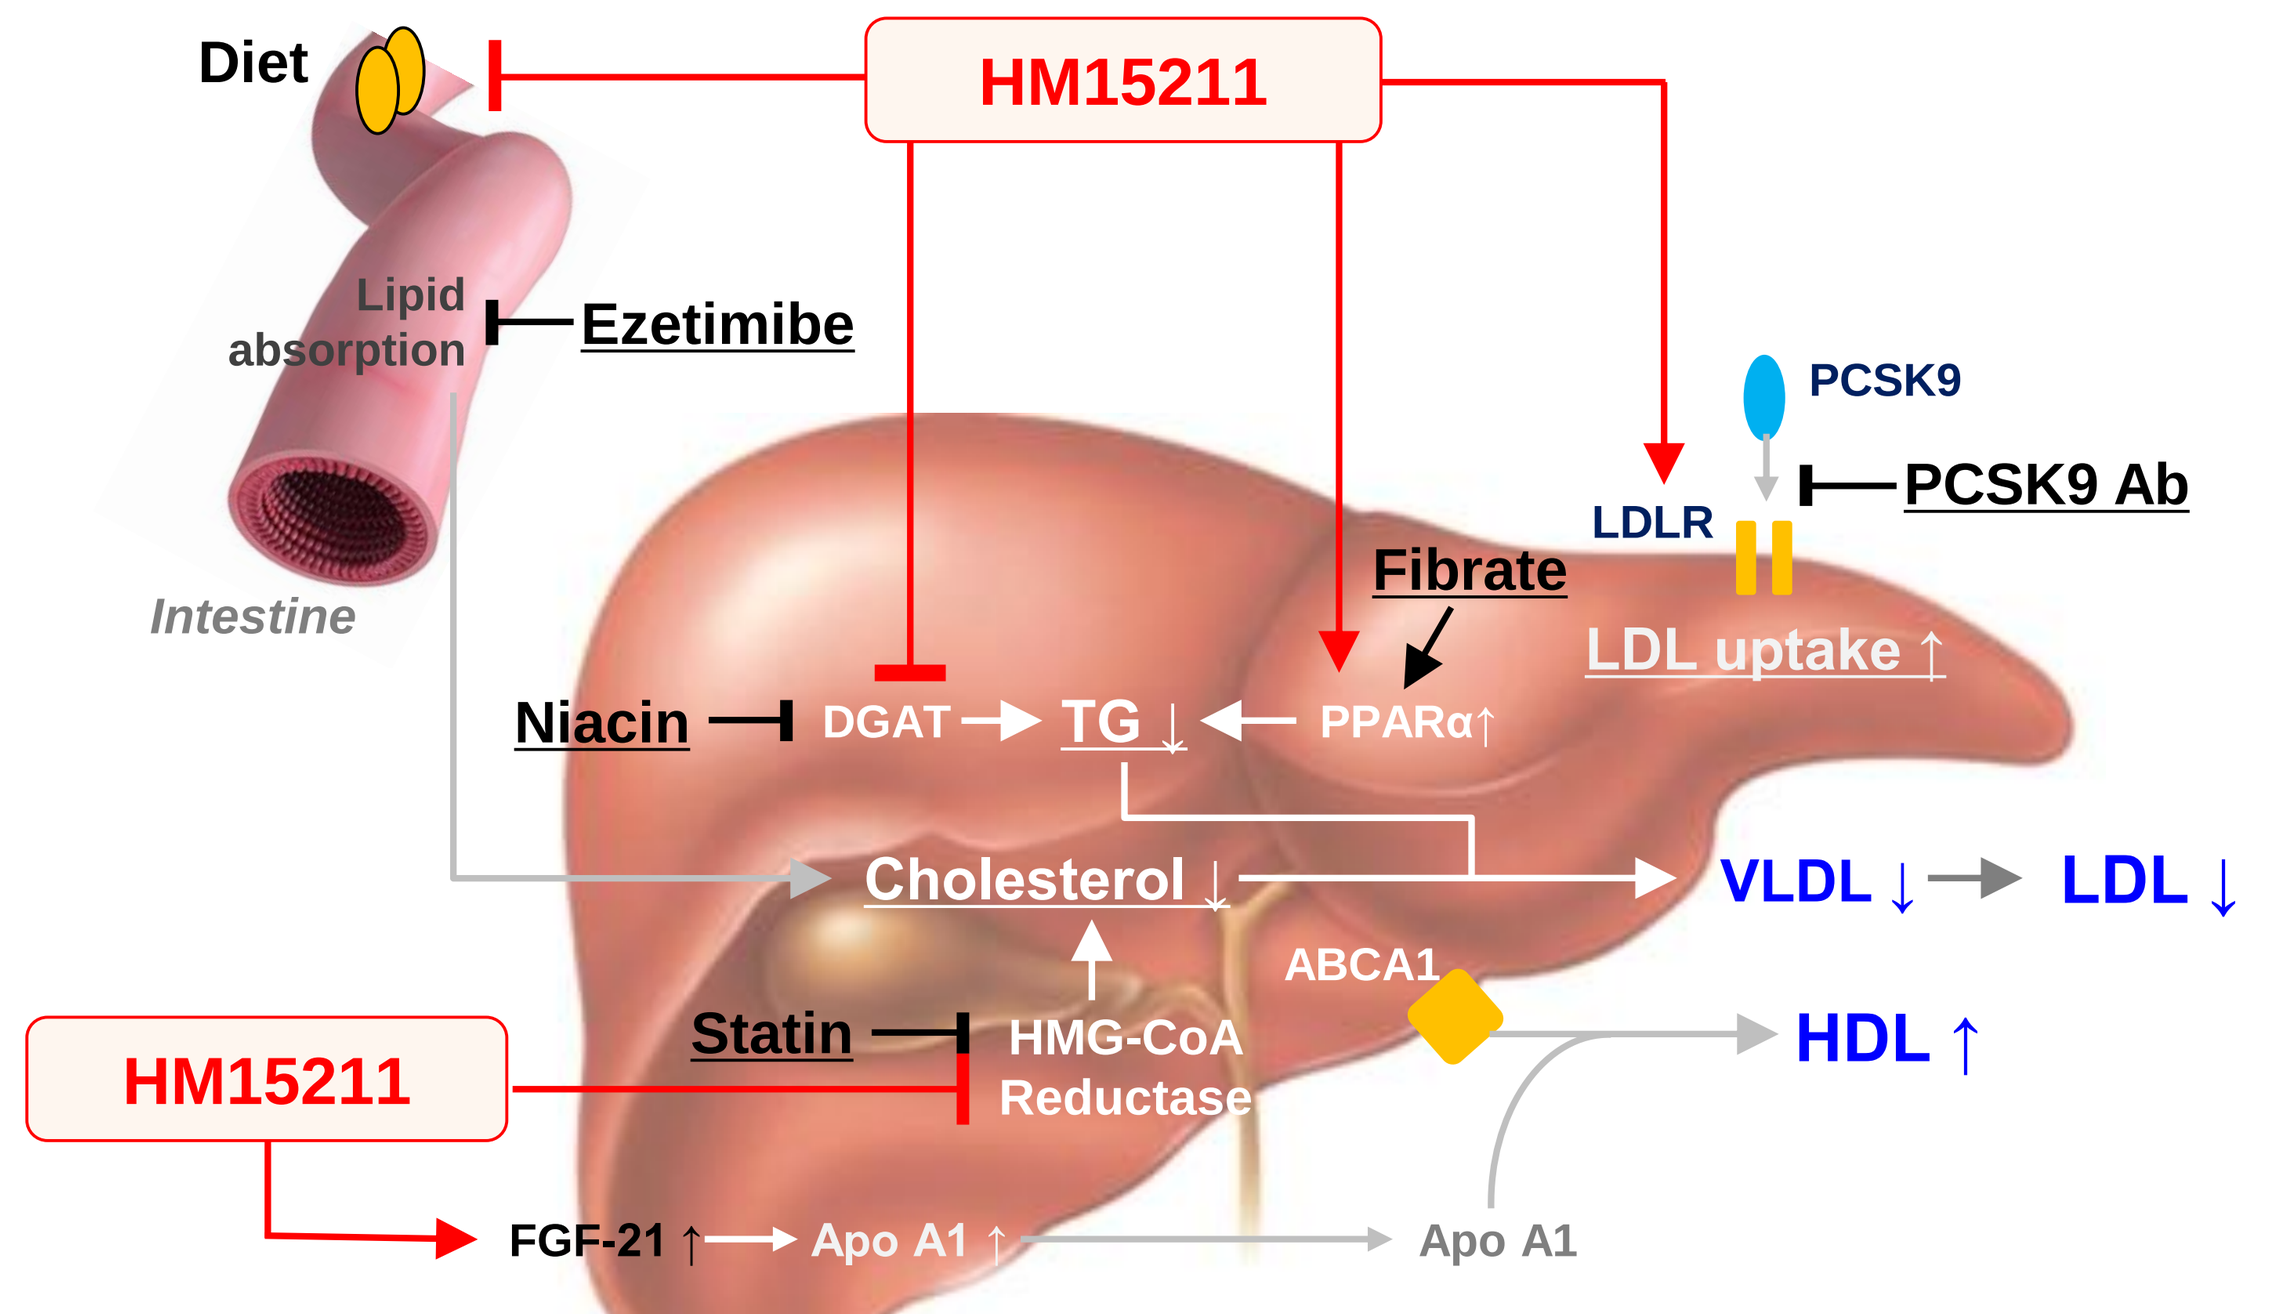

Diet
HM15211
Lipid absorption
Ezetimibe
Intestine
PCSK9
PCSK9 Ab
LDLR
Fibrate
LDL uptake ↑
Niacin
DGAT
TG ↓
PPARα↑
Cholesterol ↓
VLDL ↓
LDL ↓
ABCA1
Statin
HMG-CoA Reductase
HDL ↑
HM15211
FGF-21 ↑
Apo A1 ↑
Apo A1

Current dyslipidemia drugs possess single target. So, the efficacy was limited even by combination therapy. With GLP-1, GIP, and GCG triple-agonism, HM15211 might provide more promising lipid lowering efficacy

METHODS

- To evaluate the therapeutic efficacy in dyslipidemia, high-fat and high-fructose diet hamsters were administered with HM15211, and blood lipid profiles were monitored. For efficacy comparison, commercially available dyslipidemia drugs such as evolocumab and rosuvastatin were included. At the end of study, liver tissue samples were prepared, and protein expression of LDLR and HMGCR was further evaluated
- To evaluate the potential inhibitory effect of HM15211 on lipid absorption, oral lipid tolerance test was performed. Briefly, overnight fasted normal mice were fed with olive oil, followed by blood TG monitoring
- For *in vitro* MoA studies, cell lysates of HepG2 cells treated with HM15211 were subjected to western blot analysis (LDLR, and HMGCR) and qPCR (lipid metabolism-related genes)
- Additionally, LDL uptake and enzymatic activity of HMGCR in HepG2 cells were also determined after HM15211 treatment by using commercially available kits

Results

Lipid lowering efficacy in an animal model

Figure 1. Effect of HM15211 on blood cholesterol (CHO) in dyslipidemia hamsters (n=6)

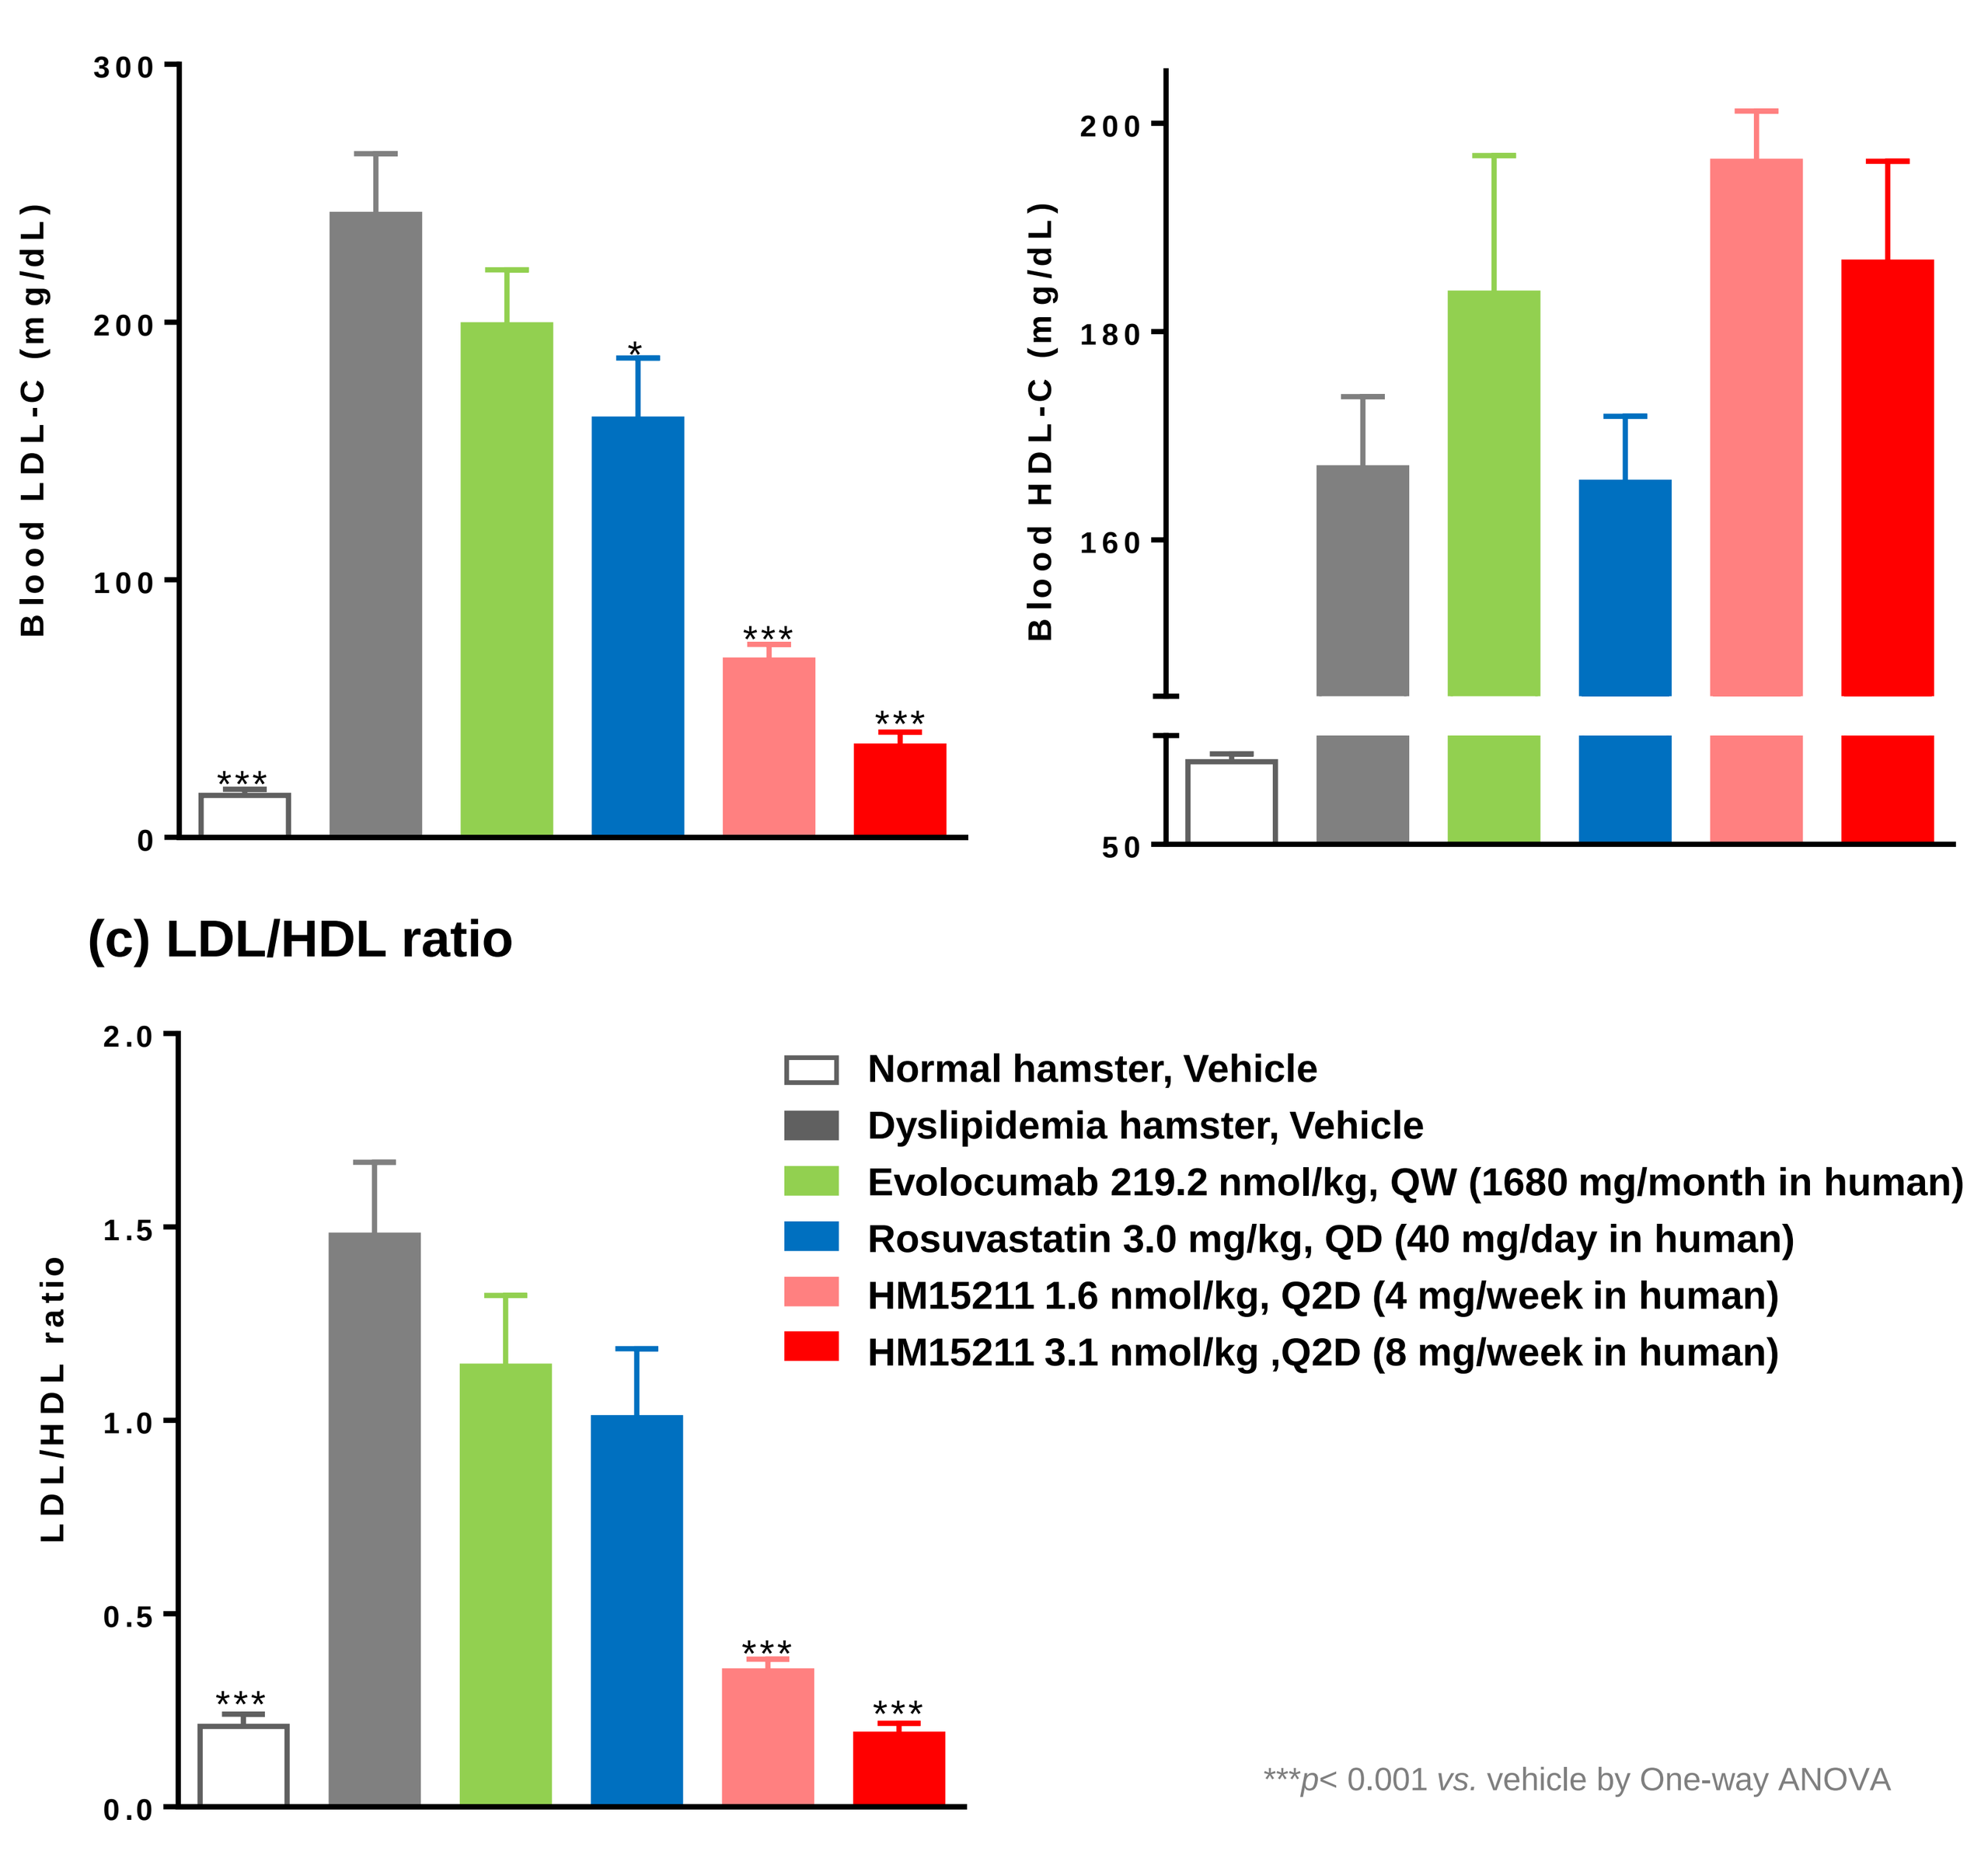

(a) LDL-C level
Blood LDL-C (mg/dL)
(b) HDL-C level
Blood HDL-C (mg/dL)
(c) LDL/HDL ratio
LDL/HDL ratio
Normal hamster, Vehicle
Dyslipidemia hamster, Vehicle
Evolocumab 219.2 nmol/kg, QW (1680 mg/month in human)
Rosuvastatin 3.0 mg/kg, QD (40 mg/day in human)
HM15211 1.6 nmol/kg, Q2D (4 mg/week in human)
HM15211 3.1 nmol/kg ,Q2D (8 mg/week in human)
***p< 0.001 vs. vehicle by One-way ANOVA

➢ In high fat- and high fructose-fed hamsters, HM15211 treatment showed superior cholesterol lowering compared to lipid lowering agents

Inhibition of lipid absorption by HM15211

Figure 2. Effect of HM15211 on lipid absorption during oil tolerance test in normal mice (n=5)

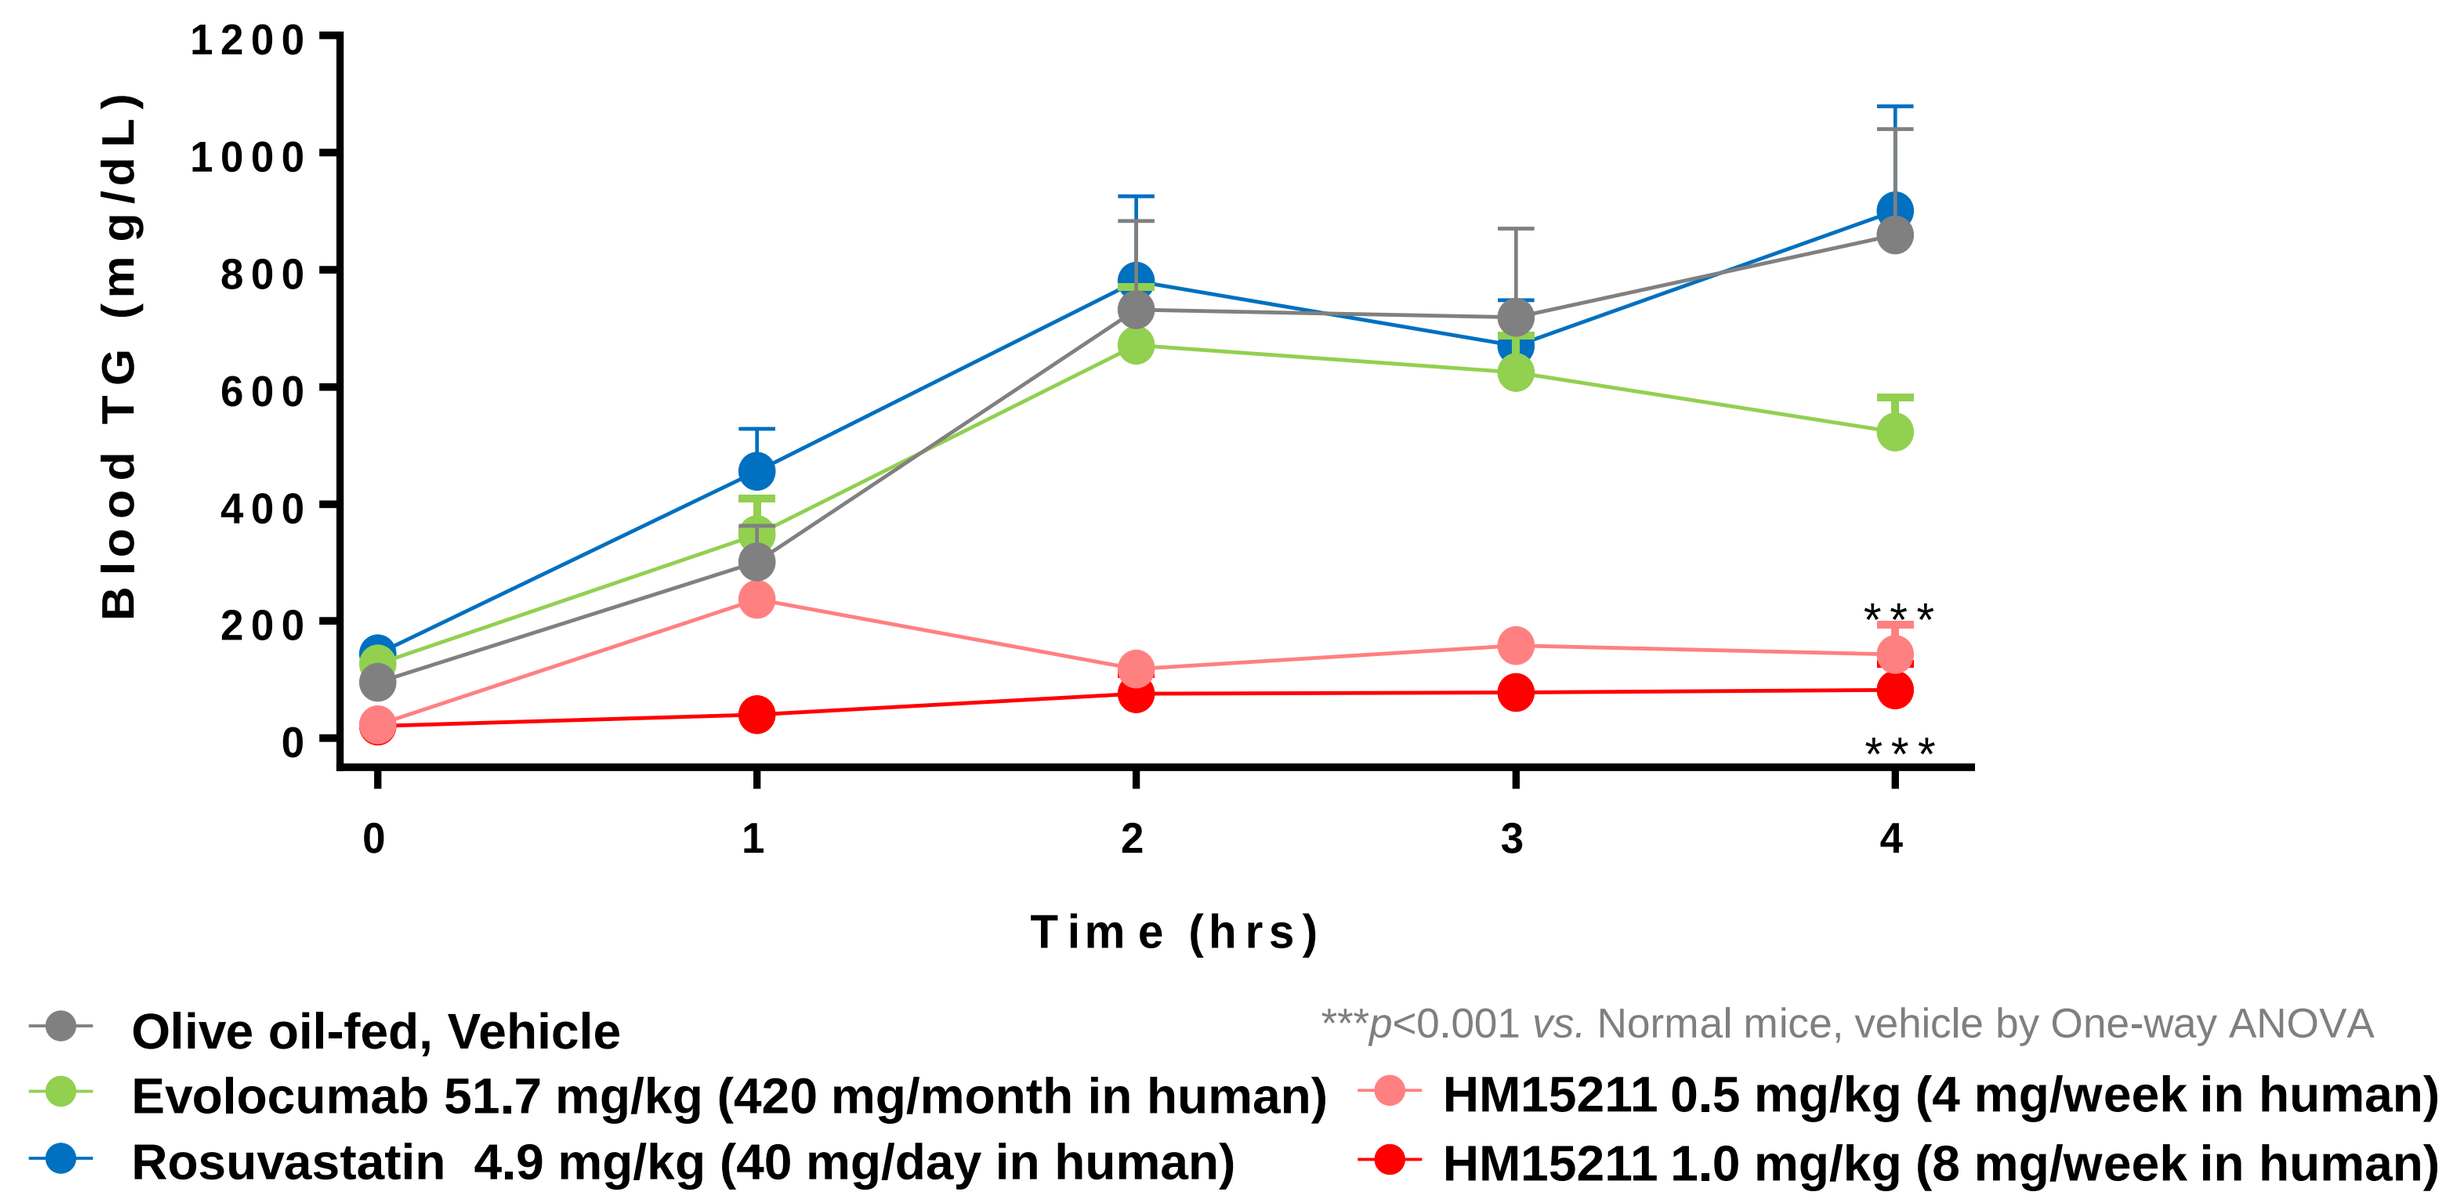

Blood TG (mg/dL)
Time (hrs)
Olive oil-fed, Vehicle
Evolocumab 51.7 mg/kg (420 mg/month in human)
Rosuvastatin 4.9 mg/kg (40 mg/day in human)
***p<0.001 vs. Normal mice, vehicle by One-way ANOVA
HM15211 0.5 mg/kg (4 mg/week in human)
HM15211 1.0 mg/kg (8 mg/week in human)

➢ Blood TG was significantly reduced in HM15211 groups after olive oil challenge, demonstrating the inhibitory effect of HM15211 on lipid absorption

Enhanced LDL clearance by HM15211

Figure 3. Effect of HM15211 on hepatic LDLR expression and LDL uptake

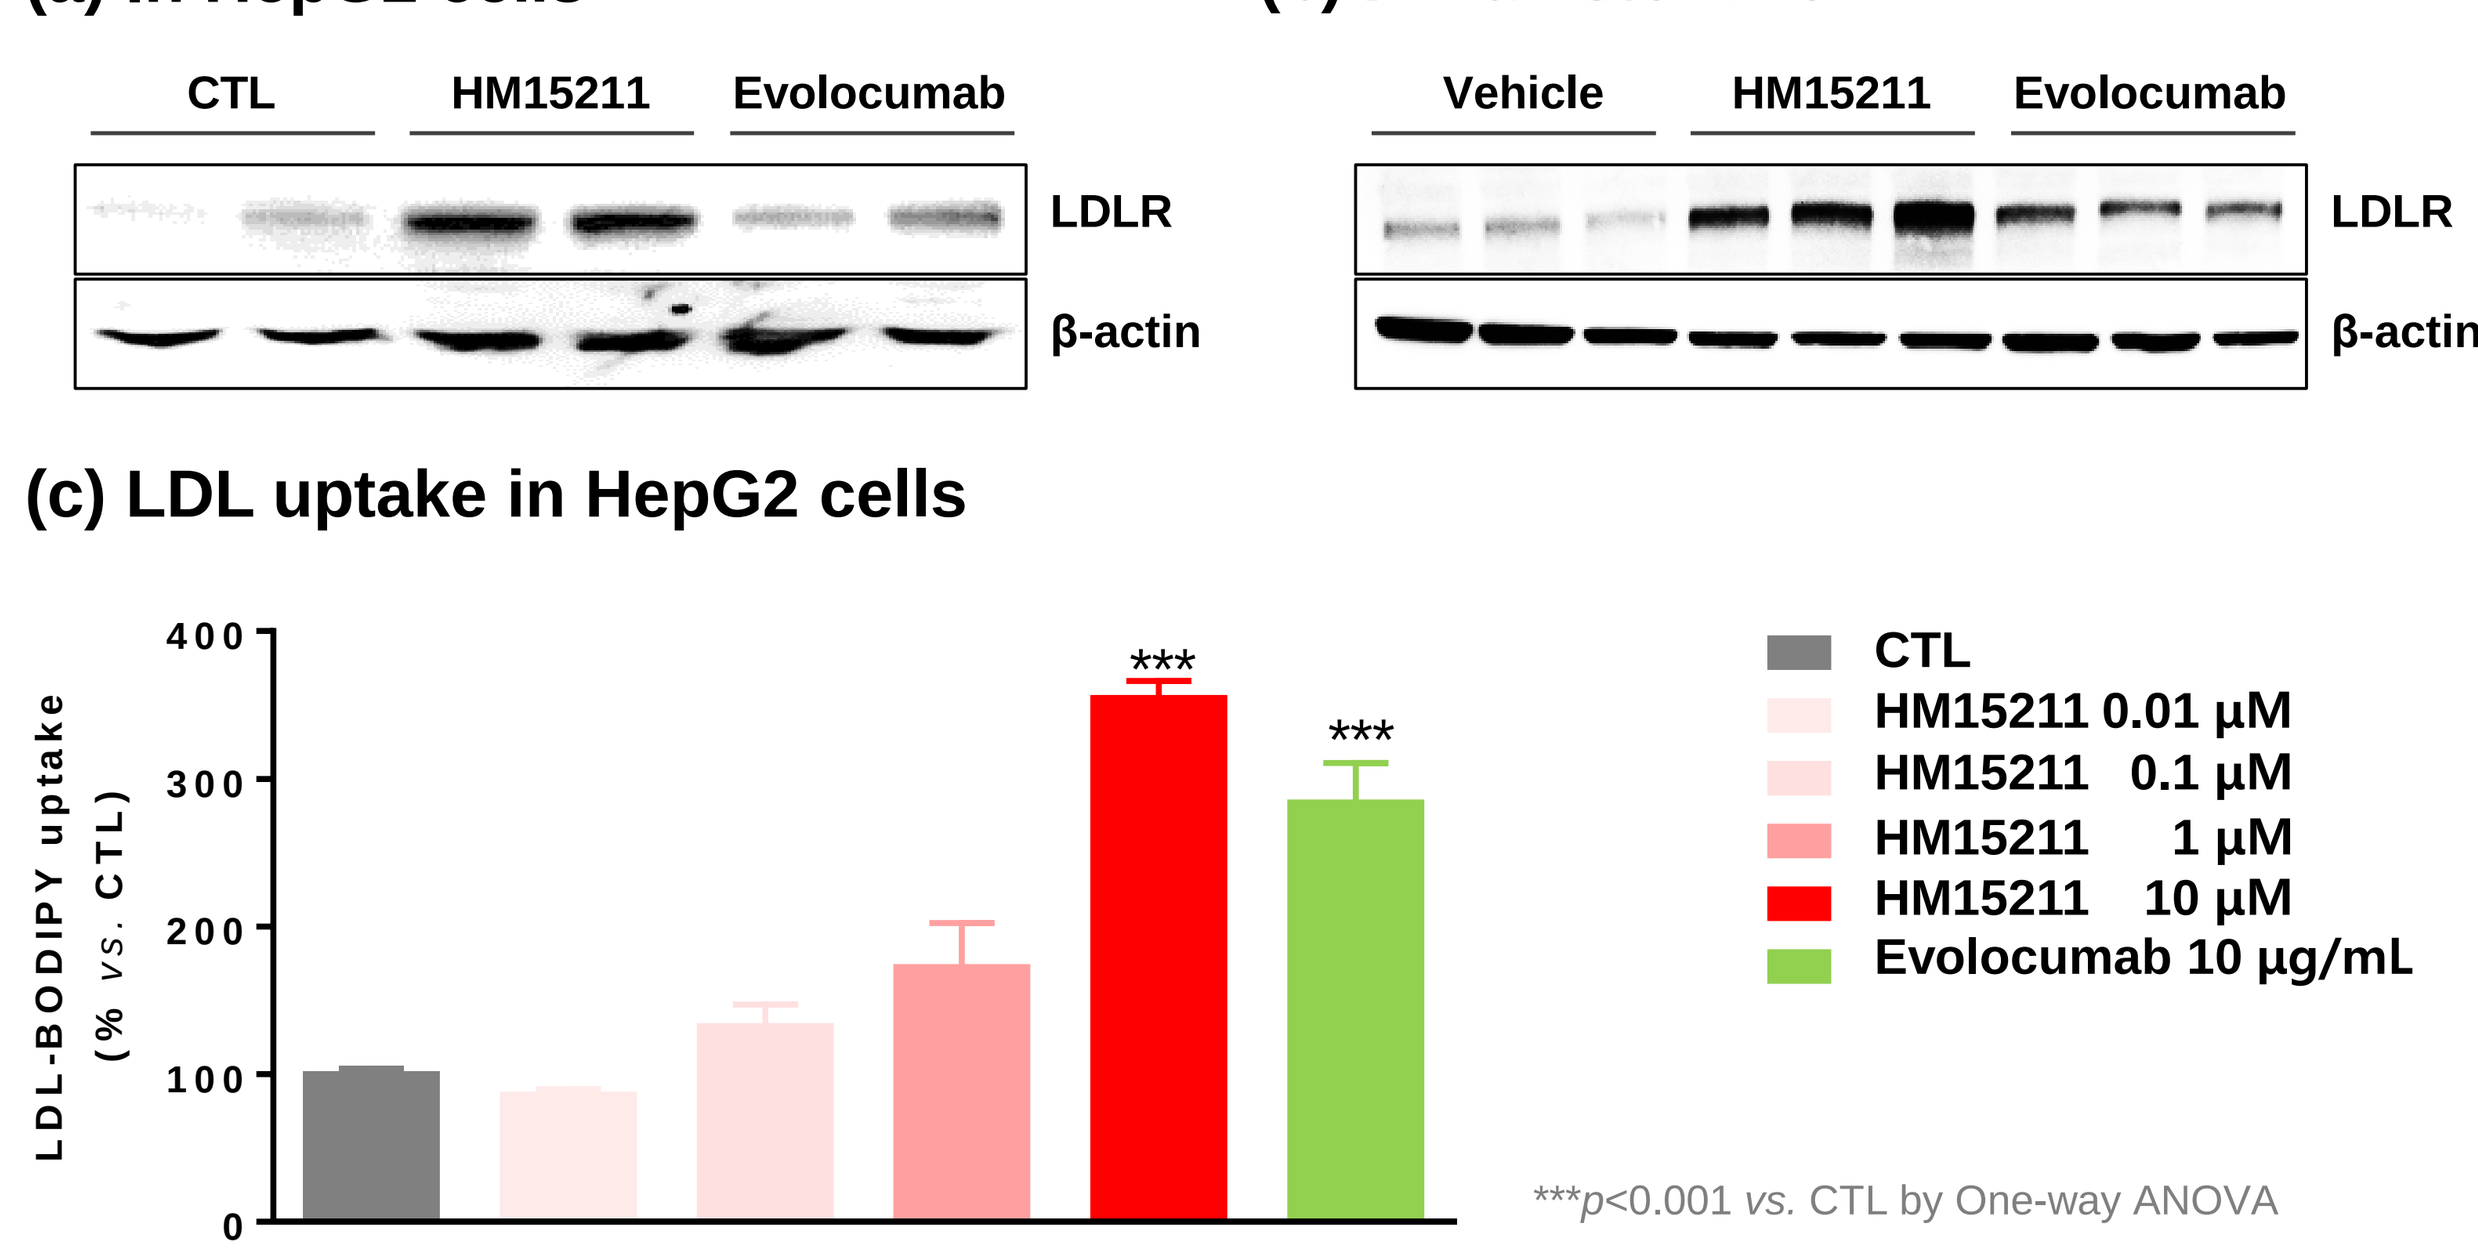

(a) In HepG2 cells
CTL
HM15211
Evolocumab
LDLR
β-actin
(b) In hamster liver
Vehicle
HM15211
Evolocumab
LDLR
β-actin
(c) LDL uptake in HepG2 cells
LDL-BODIPY uptake (% vs. CTL)
CTL
HM15211 0.01 μM
HM15211 0.1 μM
HM15211 1 μM
HM15211 10 μM
Evolocumab 10 μg/mL
***p<0.001 vs. CTL by One-way ANOVA

➢ HM15211 treatment increased LDLR expression in HepG2 cell and liver tissue, which correlated with enhanced LDL uptake by HM15211

HMGCR§ inhibition by HM15211

Figure 4. Effect of HM15211 on hepatic HMGCR expression, and enzymatic activity

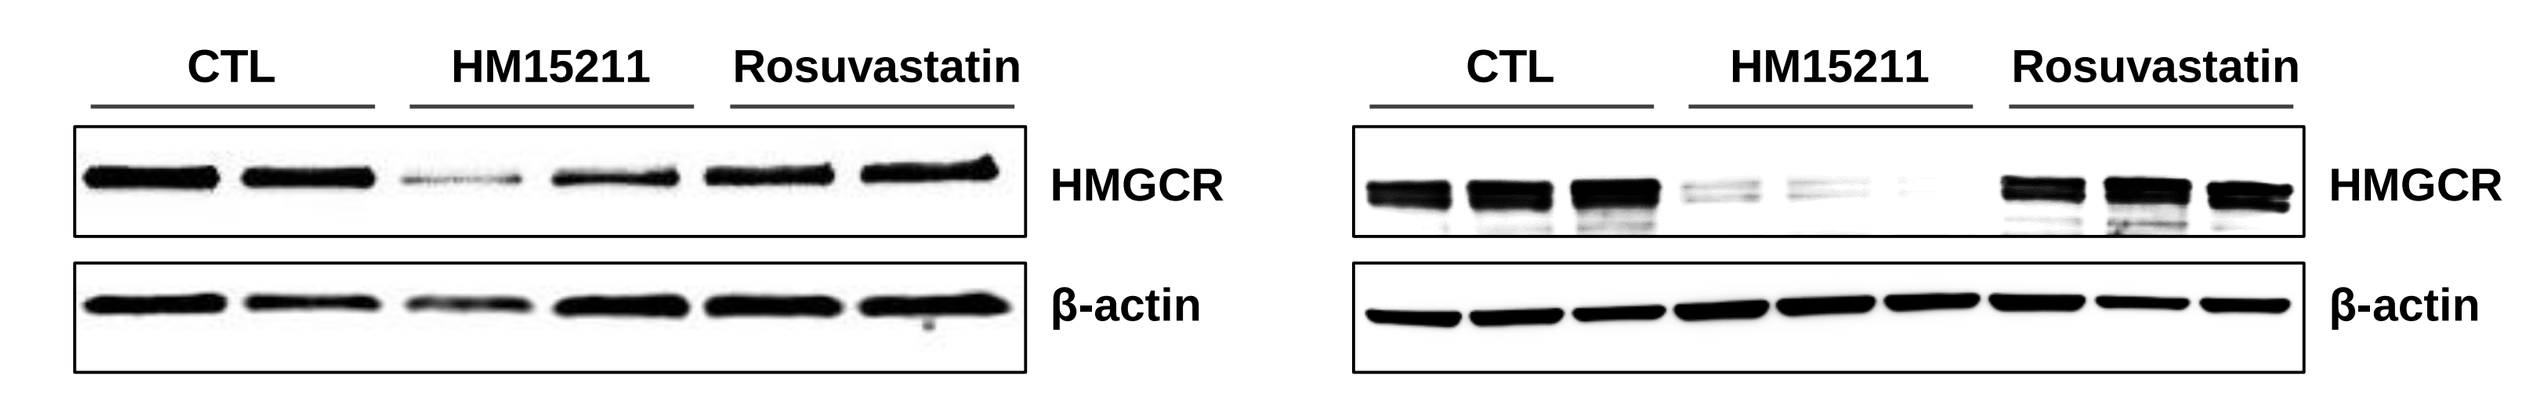

(a) In HepG2 cells
CTL
HM15211
Rosuvastatin
HMGCR
β-actin
(b) In hamster liver
CTL
HM15211
Rosuvastatin
HMGCR
β-actin

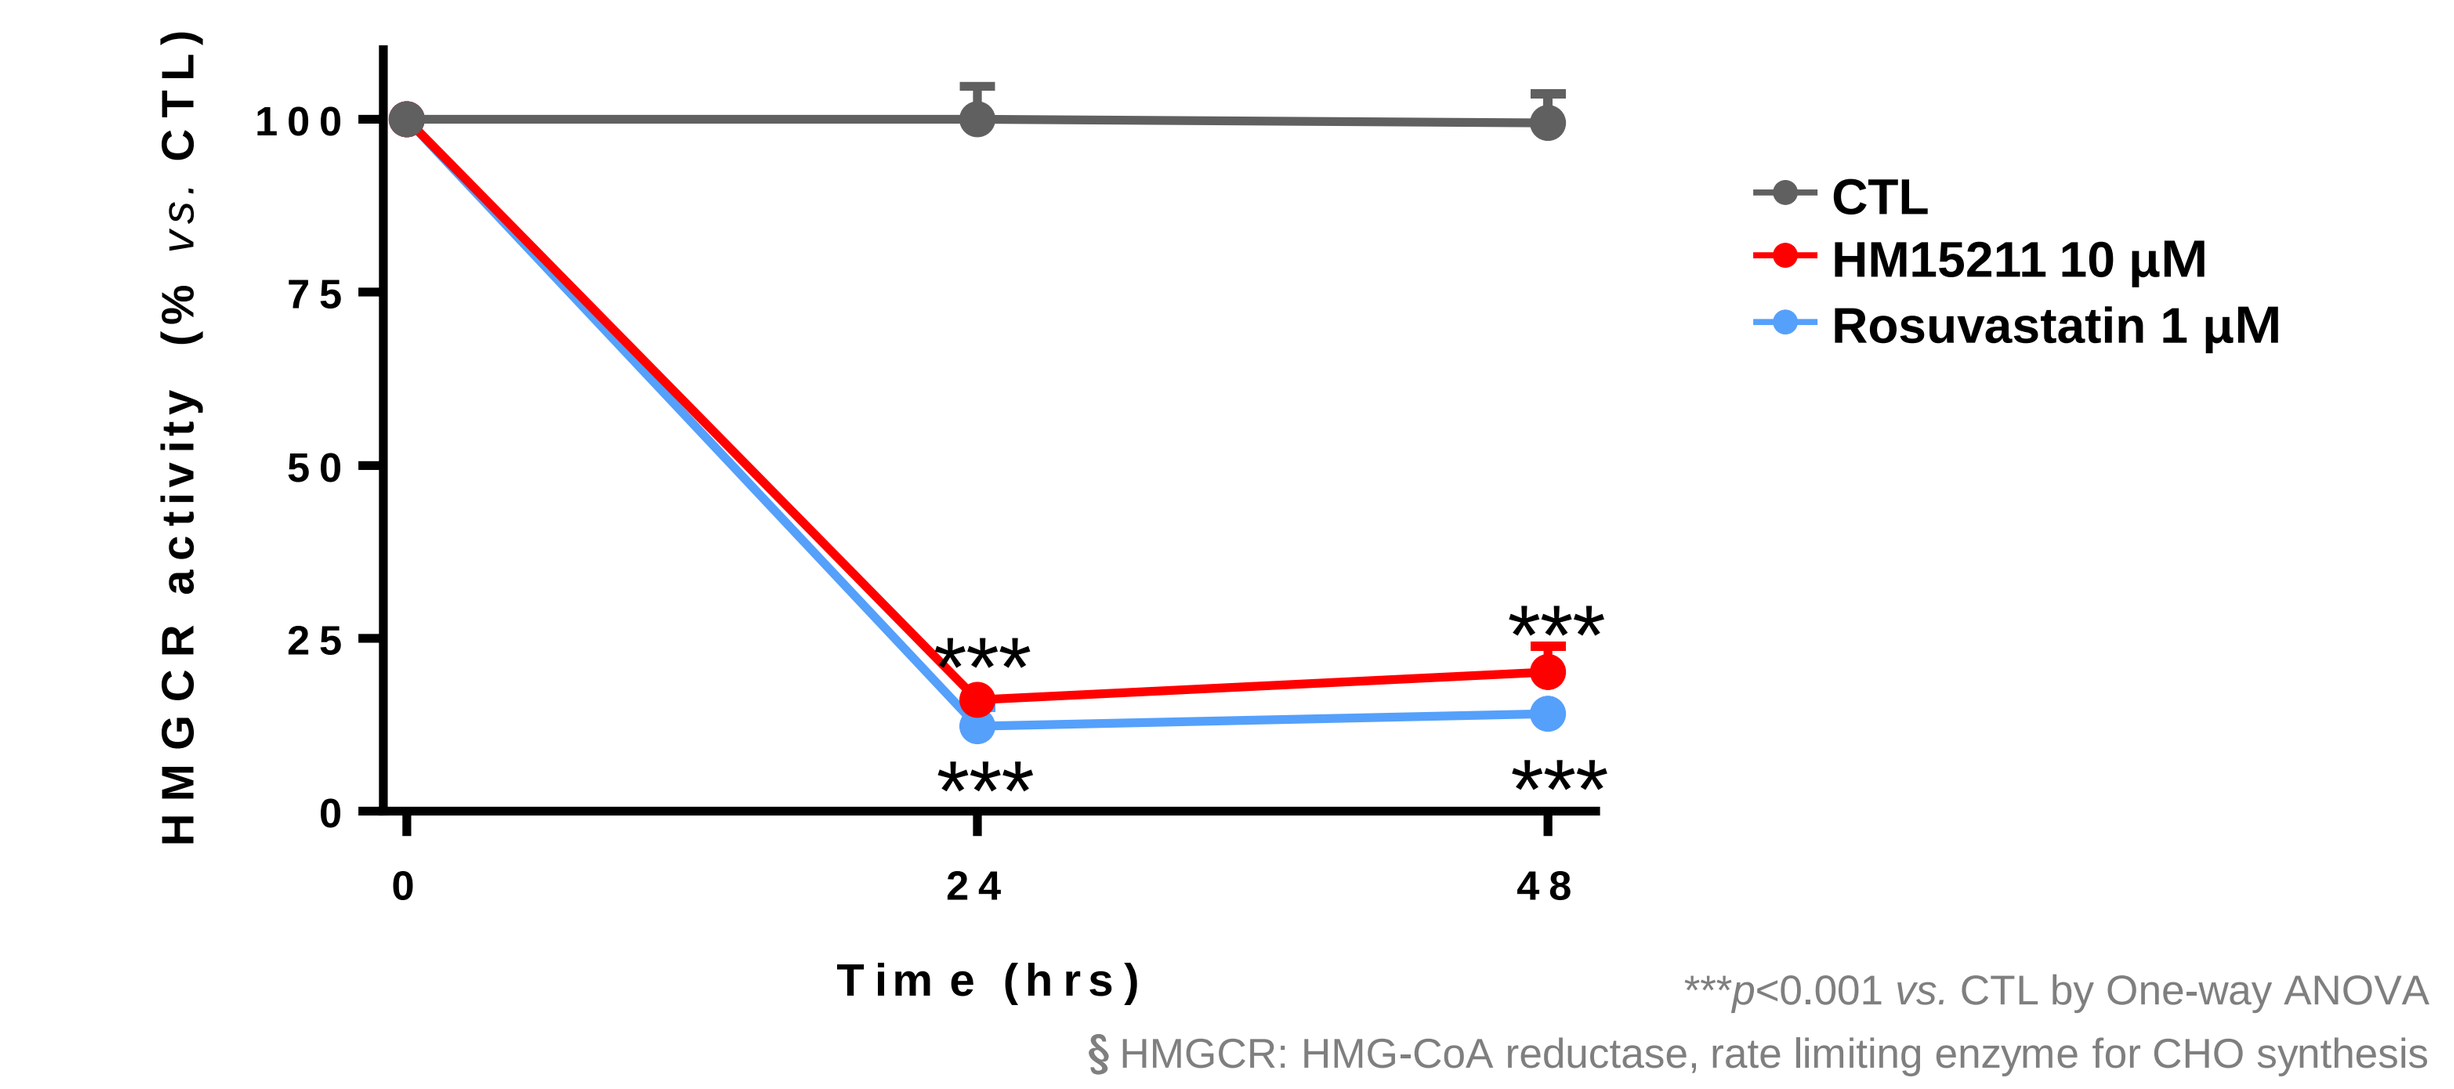

(c) HMGCR§ activity
HMGCR activity (% vs. CTL)
Time (hrs)
CTL
HM15211 10 μM
Rosuvastatin 1 μM
***p<0.001 vs. CTL by One-way ANOVA
§ HMGCR: HMG-CoA reductase, rate limiting enzyme for CHO synthesis

➢ HM15211 increased phosphorylation of HMGCR (*data not shown*), the key enzyme for CHO synthesis, followed by progressive degradation (48hr) in HepG2 cells, thereby substantially inhibiting the enzymatic activity of HMGCR

Improved FFA metabolism by HM15211

Figure 5. Effect of HM15211 on FFA metabolism related gene expression in HepG2 cells

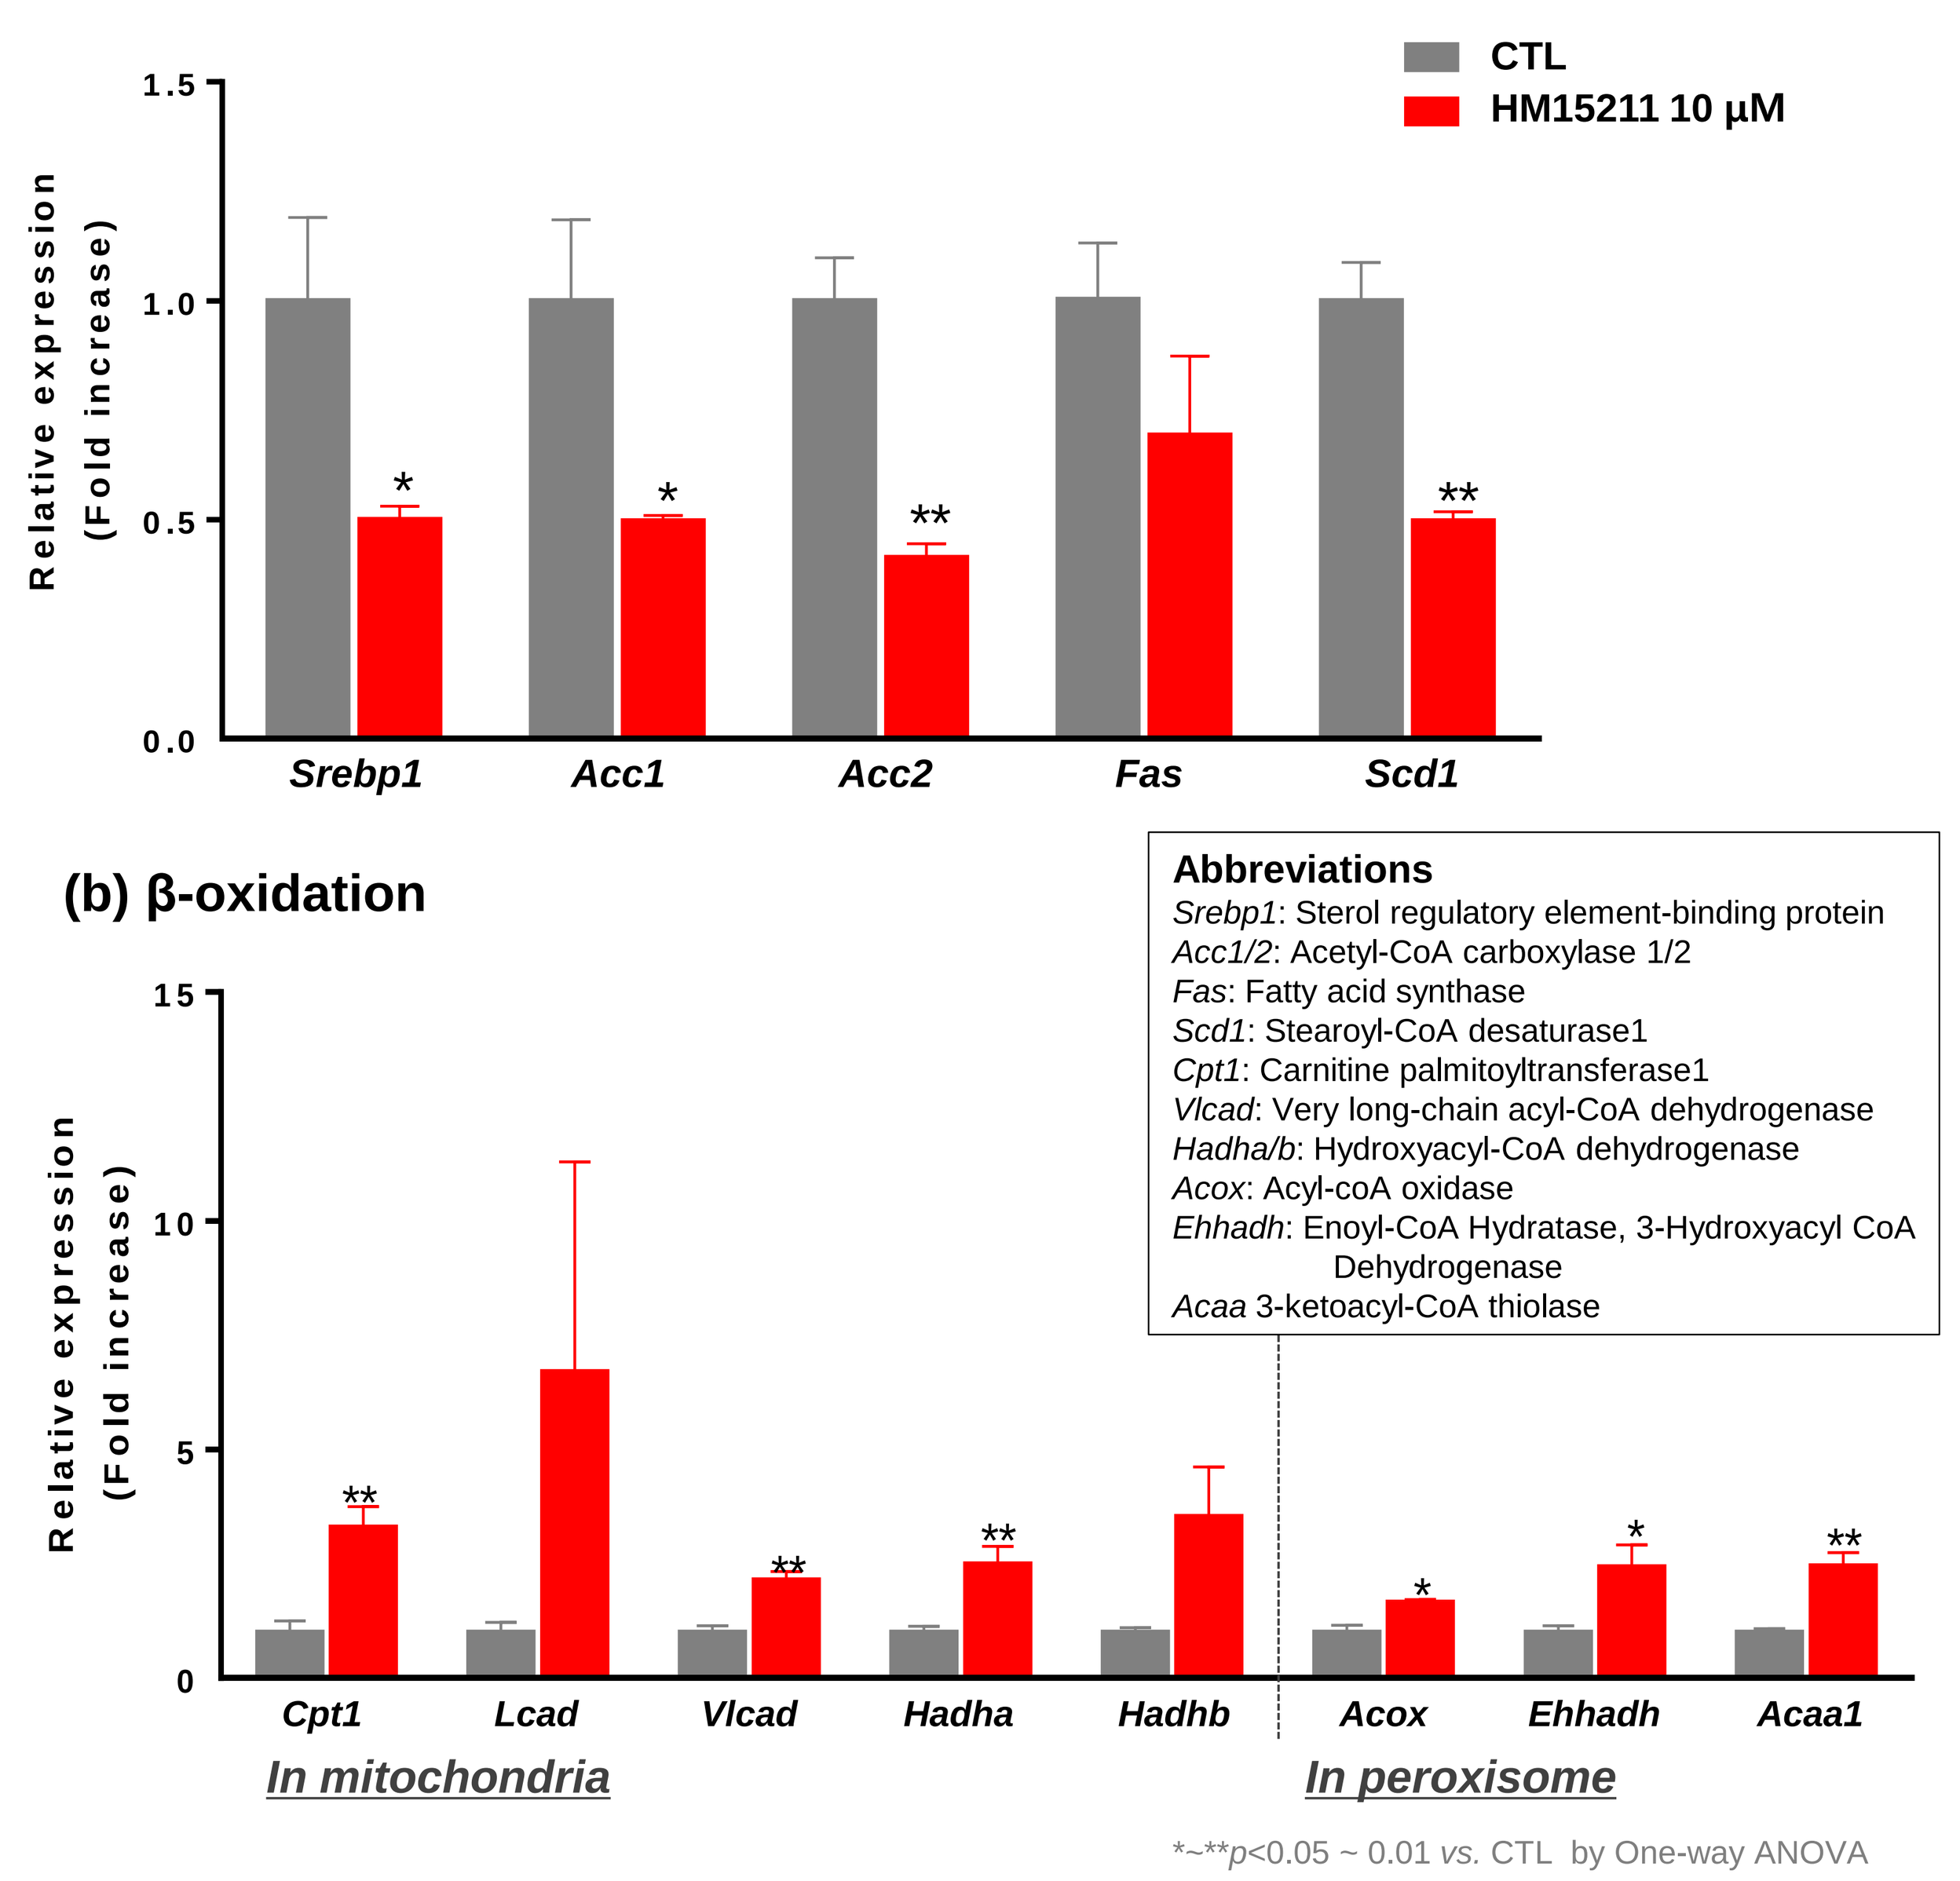

(a) De novo lipogenesis
Relative expression (Fold increase)
CTL
HM15211 10 μM
Srebp1
Acc1
Acc2
Fas
Scd1
(b) β-oxidation
Cpt1
Lcad
Vlcad
Hadha
Hadhb
Acox
Ehhadh
Acaa1
In mitochondria
In peroxisome
Abbreviations
Srebp1: Sterol regulatory element-binding protein
Acc1/2: Acetyl-CoA carboxylase 1/2
Fas: Fatty acid synthase
Scd1: Stearoyl-CoA desaturase1
Cpt1: Carnitine palmitoyltransferase1
Vlcad: Very long-chain acyl-CoA dehydrogenase
Hadha/b: Hydroxyacyl-CoA dehydrogenase
Acox: Acyl-coA oxidase
Ehhadh: Enoyl-CoA Hydratase, 3-Hydroxyacyl CoA Dehydrogenase
Acaa 3-ketoacyl-CoA thiolase
*~**p<0.05 ~ 0.01 vs. CTL by One-way ANOVA

➢ HM15211 reduced and increased the expression of genes involved in FFA synthesis and β-oxidation in HepG2 cells, suggesting favorable changes in lipid metabolism

Increased HDL synthesis markers by HM1521

Figure 6. Effect of HM15211 on hepatic ApoA1 and ABCA1§ expression

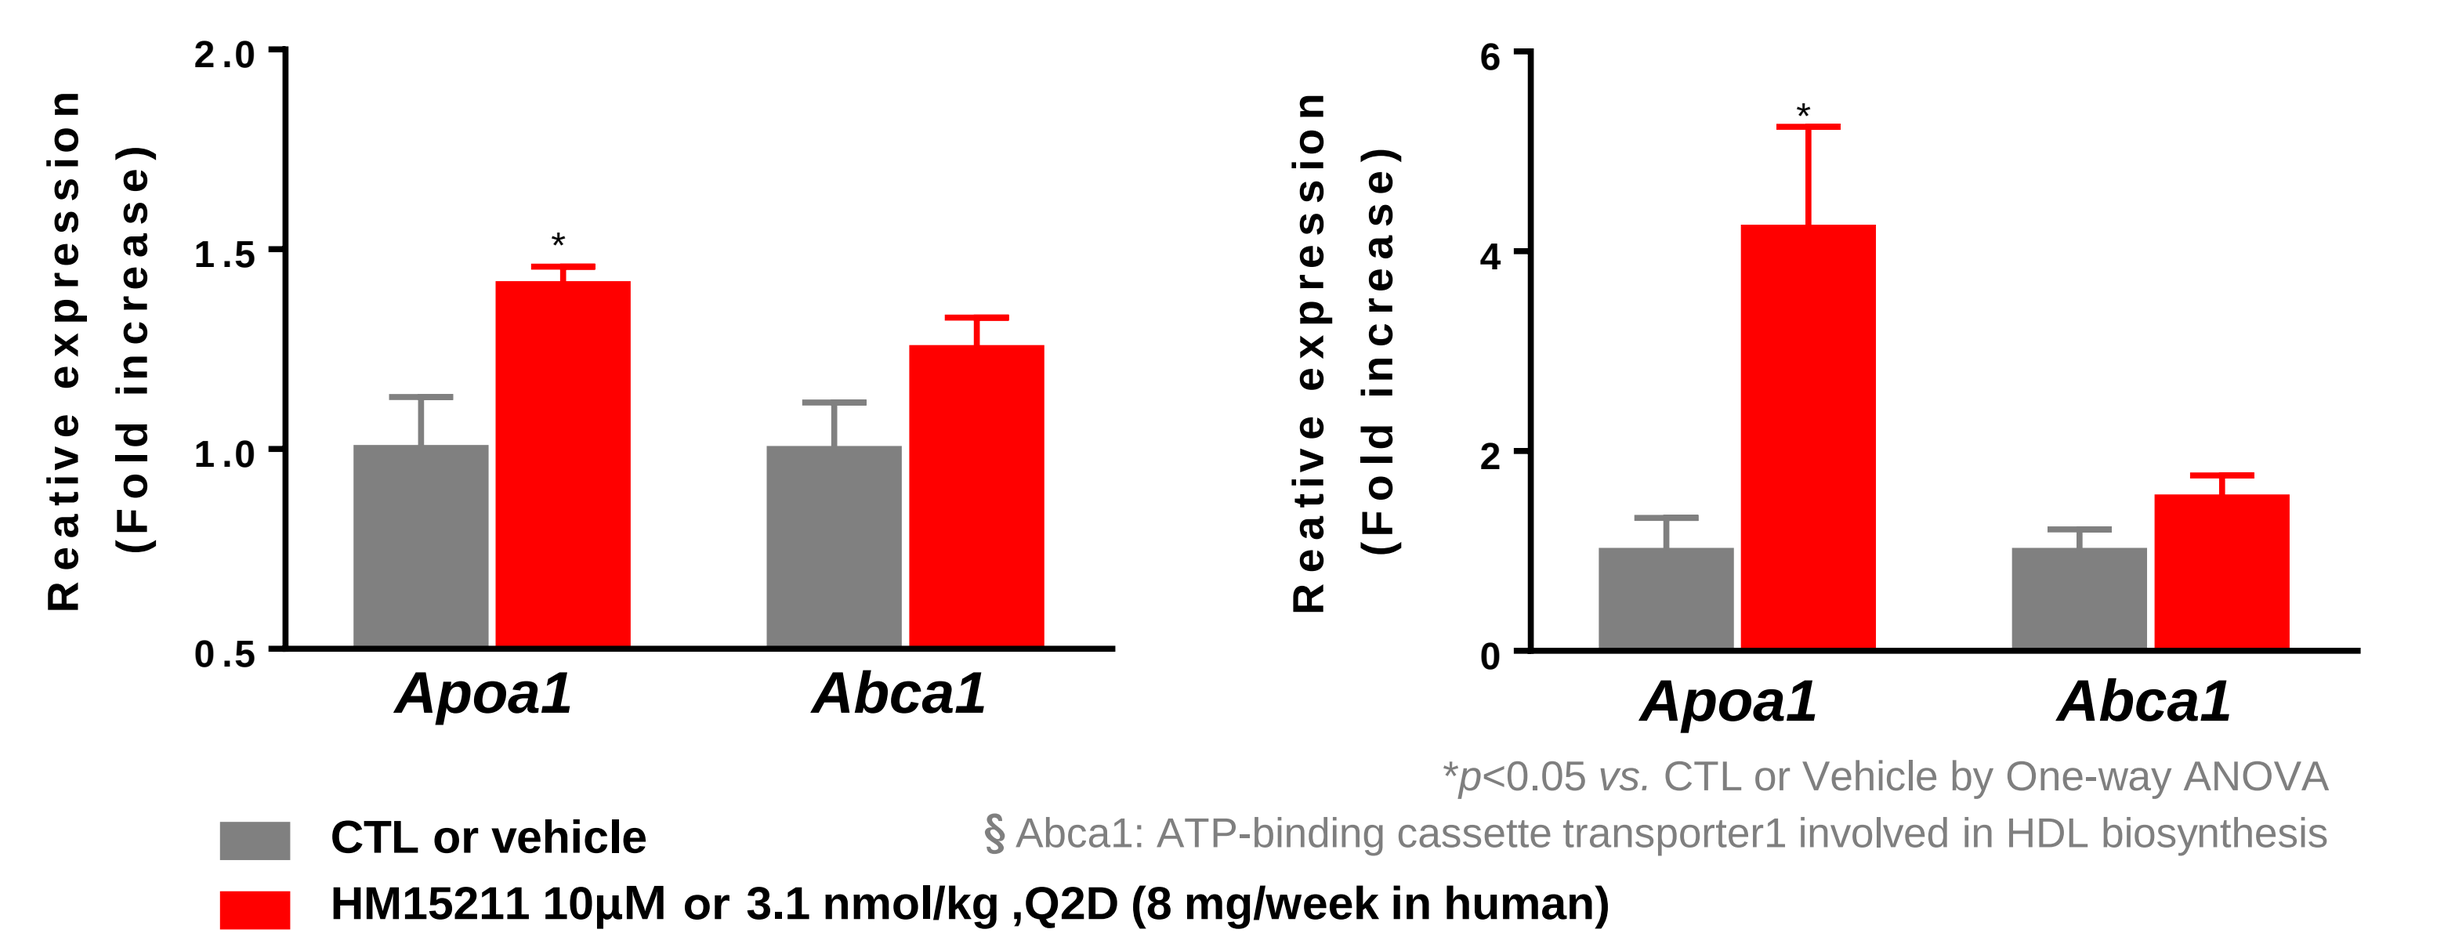

(a) In HepG2 cells
(b) In hamster liver
Reative expression (Fold increase)
Apoa1
Abca1
*p<0.05 vs. CTL or Vehicle by One-way ANOVA
CTL or vehicle
§ Abca1: ATP-binding cassette transporter1 involved in HDL biosynthesis
HM15211 10μM or 3.1 nmol/kg ,Q2D (8 mg/week in human)

➢ HM15211 tended to increase hepatic expression of ApoA1 and ABCA1, suggesting potential benefits of HM15211 in HDL synthesis

Figure 7. Proposed MoAs for efficient lipid lowering by HM15211

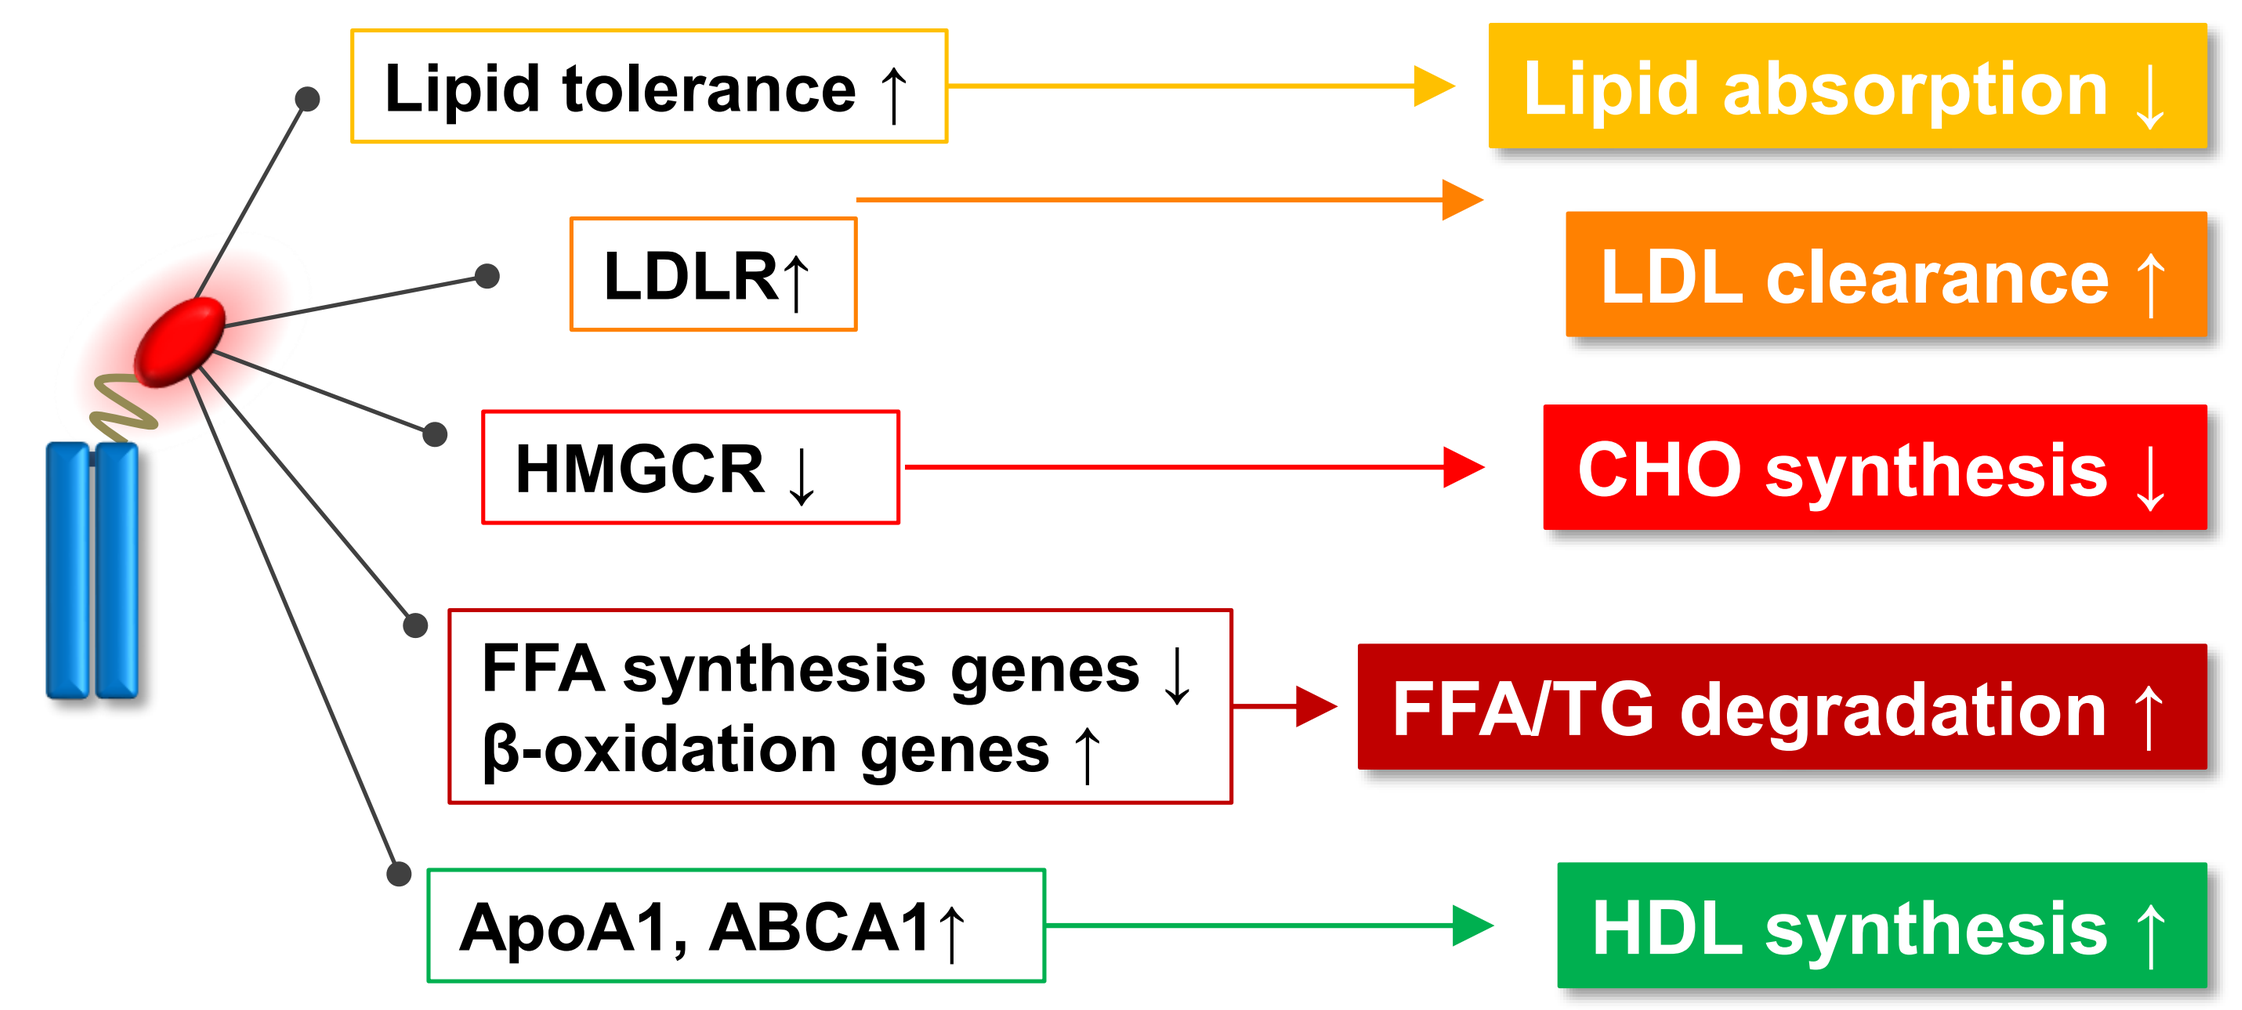

Lipid tolerance ↑
Lipid absorption ↓
LDLR↑
LDL clearance ↑
HMGCR ↓
CHO synthesis ↓
FFA synthesis genes ↓
β-oxidation genes ↑
FFA/TG degradation ↑
ApoA1, ABCA1↑
HDL synthesis ↑

CONCLUSIONS

- **In dyslipidemia hamsters, HM15211 provides greater CHO lowering than commercial dyslipidemia drugs such as evolocumab and statin**
- **In the series of mechanistic studies, responsible modes of action for this potent CHO lowering by HM15211 are elucidated as follows: (1) inhibition of lipid absorption, (2) enhanced LDL clearance, (3) inhibition of CHO synthesis, (4) improved FFA metabolism, and (5) potential for HDL synthesis**
- **In conclusion, our results suggest that HM15211 might be a good therapeutic option for dyslipidemia patients**

REFERENCES

- A Kharitonenkov *et al, Mol Metab.* 3, 221-9 (2013)
- E Osto *et al, Circulation*. 131, 871-81 (2015)
- JS Dron *et al, Arterioscler Thromb Vas Biol*. 38, 592-8 (2018)

American Diabetes Association's (ADA) 79th Scientific Sessions, San Francisco, CA, USA; June 07-11, 2019

Hanmi Pharm. Co., Ltd.